4130

Relacorilant With Nab-paclitaxel: Safety and Activity in Patients With Pancreatic Ductal Adenocarcinoma and Ovarian Cancer

Pamela N. Munster[1]; Jasgit C. Sachdev[2]; Gini F. Fleming[3]; Erkut Borazanci[2]; Jennifer Grabowsky[1]; Manish Sharma[3]; Joseph M. Custodio[4]; Andrew Greenstein[4]; Lawrence H. Lu[4]; Dat Nguyen[4]; Stacie Peacock Shepherd[4]

[1]Department of Medicine (Hematology/Oncology), University of California San Francisco, San Francisco, CA, USA; [2]HonorHealth Research Institute, Scottsdale, AZ, USA; [3]Department of Medicine, The University of Chicago, Chicago, IL, USA; [4]Corcept Therapeutics, Menlo Park, CA, USA

Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

INTRODUCTION

CORTISOL MODULATION AND CANCER BIOLOGY

- High rates of glucocorticoid receptor (GR) expression have been shown in several solid tumor types, including pancreatic, ovarian, and triple-negative breast cancer[1,2]
- Glucocorticoid (GC) mediates tumor progression via epithelial-mesenchymal transition, TGF-β induction, and gemcitabine resistance in PDAC xenografts[3]
- High GR expression correlates with lower median progression-free survival in patients with ovarian cancer[4]
- GC-mediated GR activation inhibited cell death induced by gemcitabine and cisplatin in pancreatic ductal adenocarcinoma (PDAC) xenografts and cell lines, and inhibited cell death induced by gemcitabine and carboplatin in ovarian cancer cell lines[5-7]

RELACORILANT: A SELECTIVE GR MODULATOR

- Relacorilant (formerly CORT125134) is a selective cortisol modulator that potently binds GR, inhibits GR in cells, and does not bind to the androgen receptor or the progesterone receptor (Ki >10 μM)[8]
- In vivo study in pancreatic cancer: Relacorilant similarly restored sensitivity to paclitaxel in the pancreatic cancer MIA PaCa-2 xenograft model (**Figure 1A1-1A2**).
- In vitro studies in ovarian cancer: In the OVCAR-5 cell line, relacorilant restored sensitivity to gemcitabine, paclitaxel, and oxaliplatin in the presence of GC (100 nM dexamethasone) (**Figure 1B1-1B3**). This effect was dose dependent when assessed with carboplatin (**Figure 1C**).

Figure 1. Relacorilant increases tumor cell sensitivity to chemotherapeutic agents in vitro and in vivo

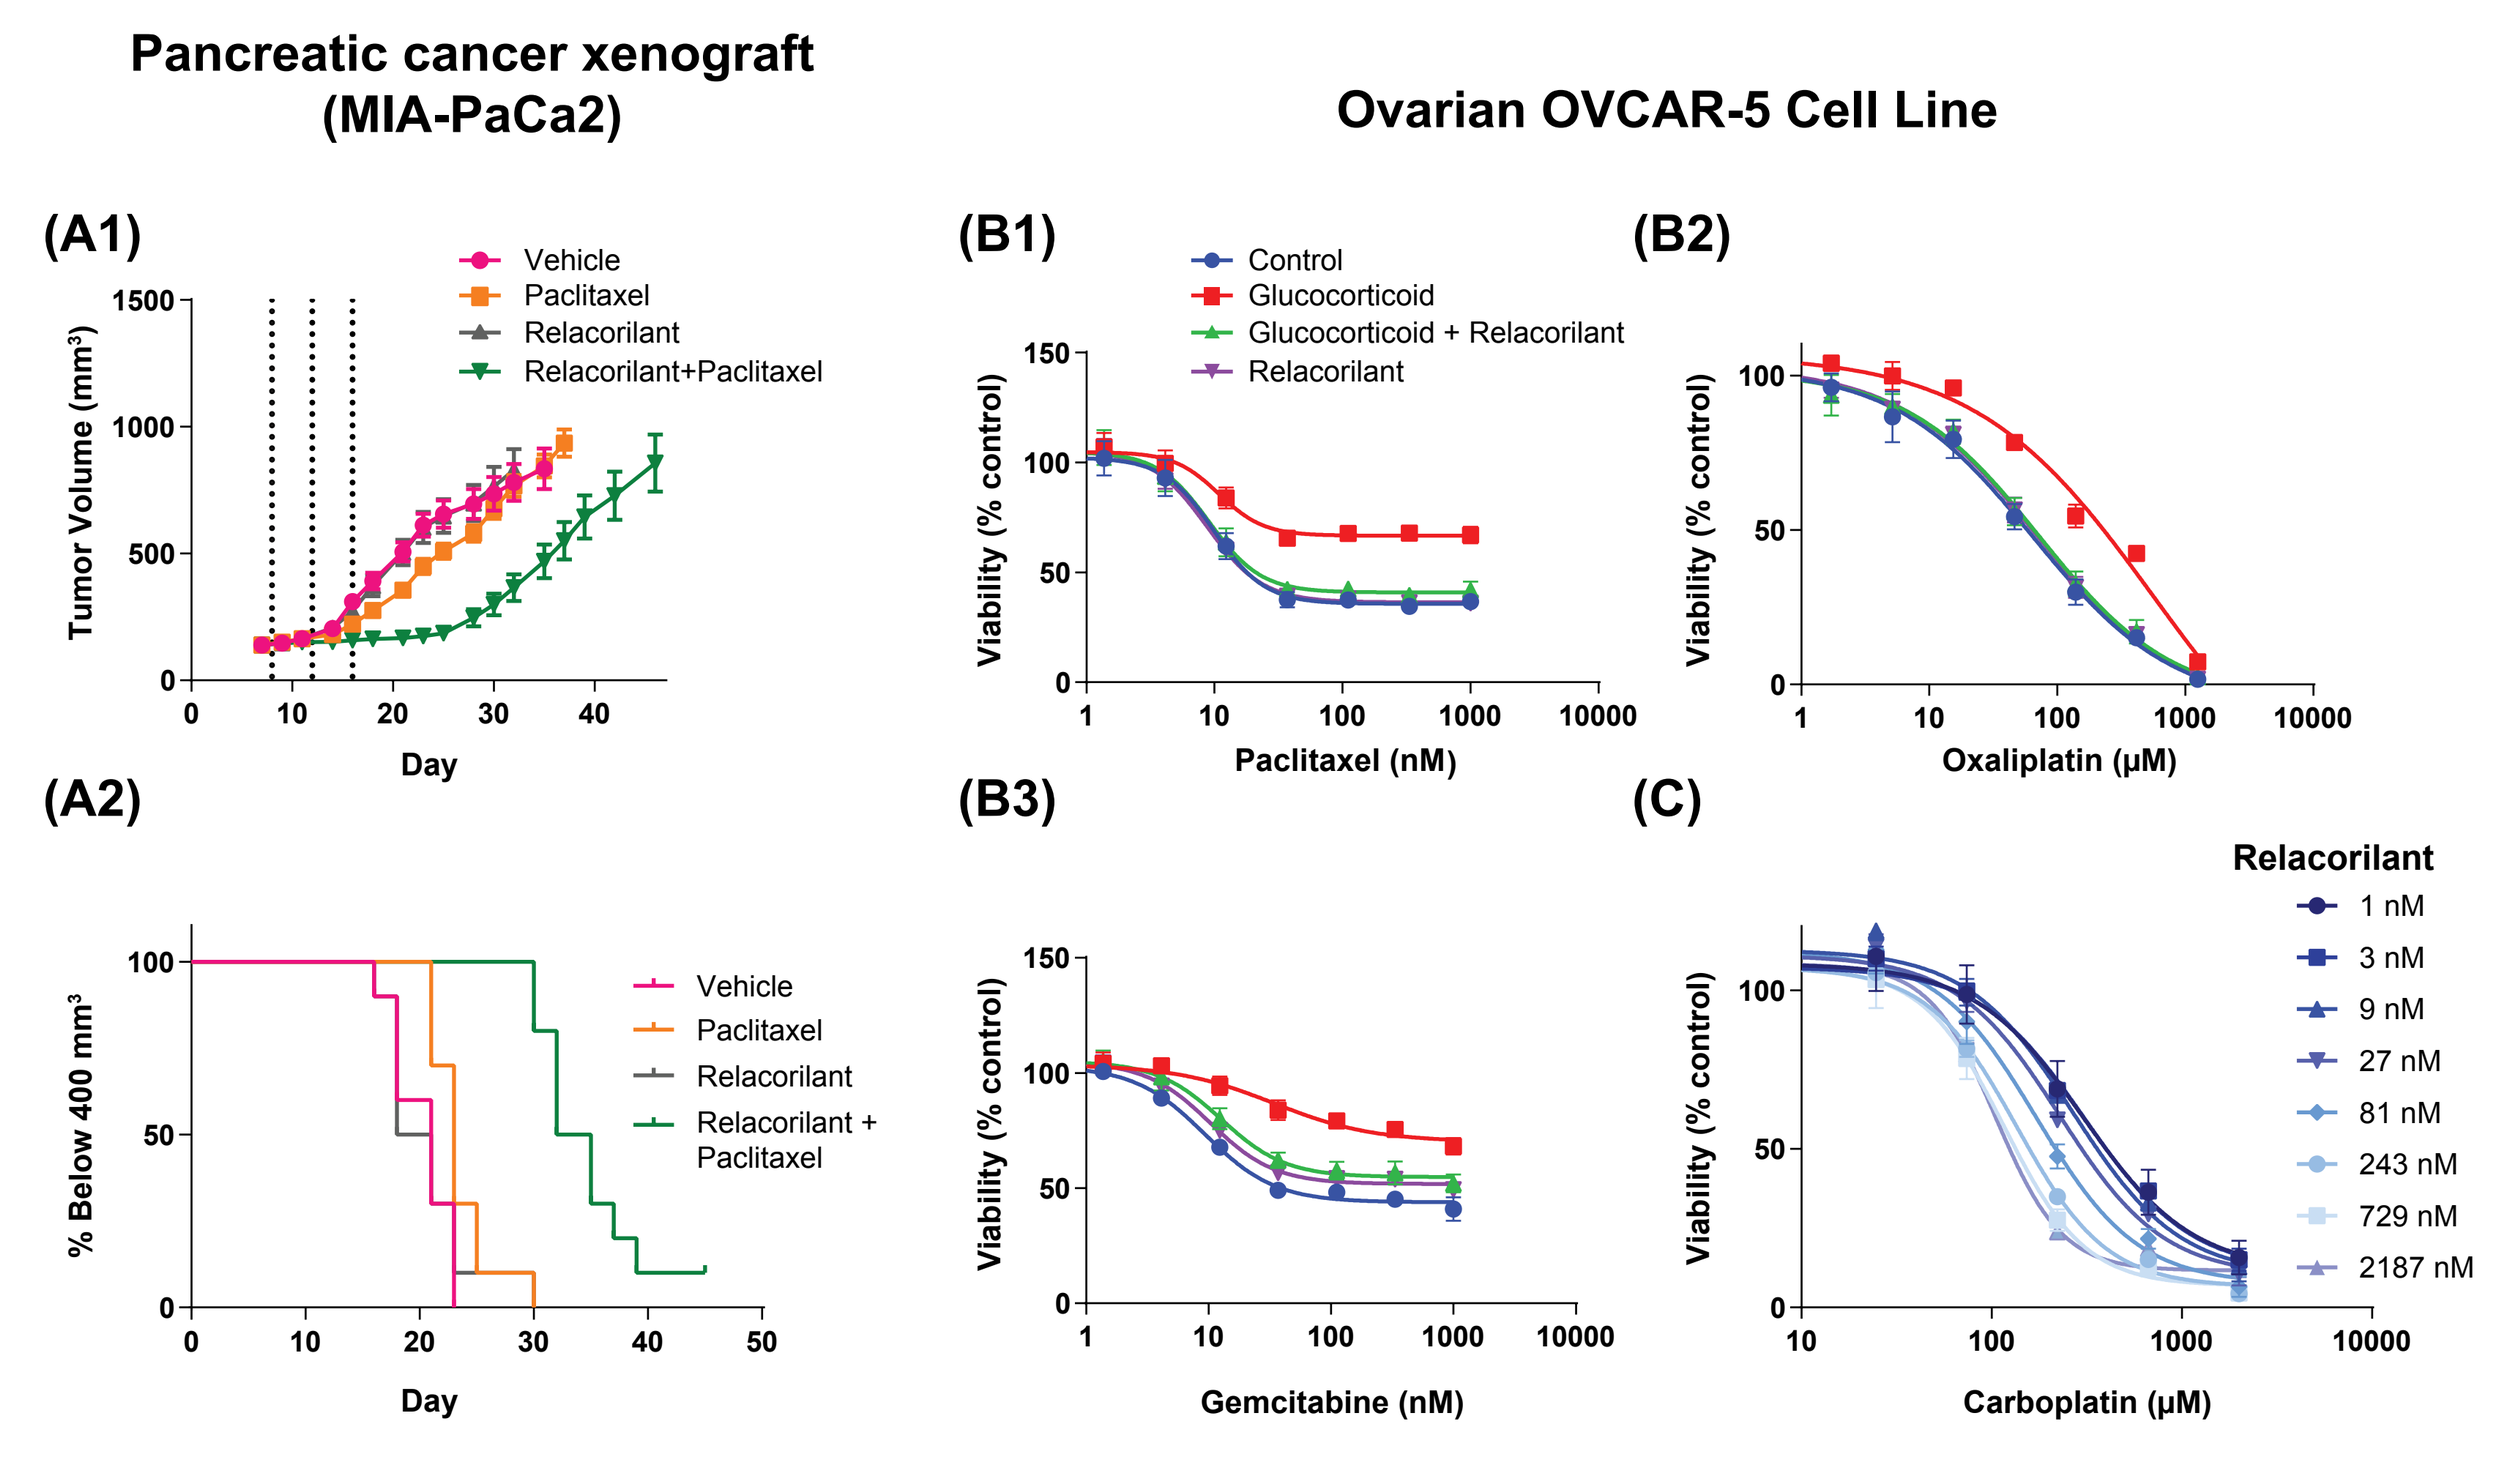

Pancreatic cancer xenograft (MIA-PaCa2)
Ovarian OVCAR-5 Cell Line
(A1)
Vehicle
Paclitaxel
Relacorilant
Relacorilant+Paclitaxel
Tumor Volume (mm³)
Day
(A2)
% Below 400 mm³
Relacorilant + Paclitaxel
(B1)
Control
Glucocorticoid
Glucocorticoid + Relacorilant
Viability (% control)
Paclitaxel (nM)
(B2)
Oxaliplatin (μM)
(B3)
Gemcitabine (nM)
(C)
Relacorilant
1 nM
3 nM
9 nM
27 nM
81 nM
243 nM
729 nM
2187 nM
Carboplatin (μM)

(A1-A2) Relacorilant sensitizes the MIA PaCa-2 xenograft to paclitaxel. 7.5 mg/kg paclitaxel dosed three times (dashed lines) was ineffective alone. Addition of relacorilant delayed tumor growth (A1, p<0.0001) and the time to progression (A2, p<0.0001) compared to paclitaxel alone. (B1-B3) Relacorilant restores chemotherapy sensitivity in vitro. Glucocorticoid (100 nM dexamethasone) reduced the half-maximal potency of paclitaxel, oxaliplatin, and gemcitabine in the OVCAR-5 cell line; relacorilant restored sensitivity to these agents in the presence of glucocorticoid. (C) Potency of carboplatin is dose-dependently increased by relacorilant in vitro. OVCAR-5 cells were grown in the presence of glucocorticoid. Carboplatin was titrated against increasing amounts of relacorilant.

METHODS

STUDY DESIGN

- In this ongoing study, patients were enrolled in a standard "3+3" dose escalation design to evaluate two dosing schedules for relacorilant (daily continuously vs. daily intermittently on day before, day of, and day after nab-paclitaxel [nab-pac] administration)
- Eligible patients had advanced or metastatic solid tumors, an Eastern Cooperative Oncology Group (ECOG) Performance Status (PS) of 0-1, and received up to 3 prior lines of cytotoxic or myelosuppressive therapy. Previous nab-pac was allowed. Patients received relacorilant (100, 150, or 200 mg) administered continuously or intermittently; nab-pac (60, 80, or 100 mg/m²) was dosed weekly for 3 of 4 weeks of a 28-day cycle.
- Due to an early signal of efficacy in patients with PDAC, tumor specific cohorts were added at each dose level to further assess the feasibility of the regimen in patients with refractory PDAC

ASSESSMENTS

- Safety was assessed in all patients who had received at least a single dose of relacorilant. The response-evaluable population included patients who had at least one post-baseline tumor assessment.
- Tumor assessments were performed at screening and every 6-8 wks from Cycle 1 Day 1 with confirmation of tumor response performed within no less than 4 weeks later and assessed per RECIST v1.1
- GR-regulated gene expression of whole blood samples were collected in PAXgene tubes (PreAnalytiX) at baseline and Cycle 1 Day 15. RNA was extracted and quantified using a custom NanoString probe set (NeoGenomics).

RESULTS

BASELINE CHARACTERISTICS AND DEMOGRAPHICS

- In this interim assessment of an ongoing study, 68 patients received ≥1 dose of relacorilant (**Table 1**), with the most common tumor types being PDAC (n=33) and ovarian cancer (n=14)

Table 1. Baseline characteristics and demographics

	PDAC[a] (n=33)	Ovarian cancer[a] (n=14)	Other solid tumors[b] (n=20)
Age (years), mean (SD)	62.8 (10.1)	53.1 (14.1)	60.3 (14.3)
Gender, n (%)			
Male	16 (48.5)	0	3 (15.0)
Female	17 (51.5)	14 (100)	17 (85.0)
Race, n (%)			
Asian	1 (3.0)	1 (7.1)	3 (15.0)
Black	1 (3.0)	2 (14.3)	3 (15.0)
White	29 (87.9)	11 (78.6)	13 (65.0)
Other	2 (6.1)	0	1 (5.0)
Ethnicity, n (%)			
Hispanic or Latino	4 (12.1)	1 (7.1)	2 (10.0)
Not Hispanic or Latino	29 (87.9)	13 (92.9)	18 (90.0)
No. of prior therapies, median (range)[c]	3.0 (1-5)	4.0 (2-6)	3.5 (1-8)
Received prior taxane, n (%)	25 (75.8)	14 (100)	12 (60.0)

PDAC, pancreatic ductal adenocarcinoma; SD, standard deviation.
[a]PDAC includes all pancreatic cancer patients except acinar subtype. Ovarian cancer includes all ovarian cancer patients, including primary peritoneal carcinoma of Mullerian origin.
[b]One patient in the other solid tumors subgroup was not included in this analysis due to missing tumor information.
[c]Number of distinct cancer therapy regimens.

SAFETY AND DETERMINATION OF RP2D

- The most common treatment-emergent Grade ≥3 adverse events were neutropenia, abdominal pain, anemia, hyponatremia, hypophosphatemia, and vomiting (**Table 2**)
- Neutropenia with nab-pac dose delay >7 days and febrile neutropenia have been the most common dose-limiting toxicities. Due to dose-limiting neutropenia, primary prophylaxis with granulocyte colony-stimulating factor became mandatory in later cohorts.

Table 2. Grade ≥3 adverse events occurring in ≥2 patients

Adverse event, n (%)	Continuous-dosing regimen (n=49)	Intermittent-dosing regimen (n=19)	Overall (N=68)
Patients Reporting at Least One Grade ≥3 Adverse Event	33 (67.4)	13 (68.4)	46 (67.7)
Neutropenia	7 (14.3)	6 (31.6)	13 (19.1)
Abdominal pain	4 (8.2)	1 (5.3)	5 (7.4)
Anemia	3 (6.1)	2 (10.5)	5 (7.4)
Hyponatremia	3 (6.1)	1 (5.3)	4 (5.9)
Hypophosphatemia	3 (6.1)	1 (5.3)	4 (5.9)
Vomiting	2 (4.1)	2 (10.5)	4 (5.9)
Diarrhea	2 (4.1)	1 (5.3)	3 (4.4)
Febrile neutropenia	3 (6.1)	0	3 (4.4)
Hypokalemia	2 (4.1)	1 (5.3)	3 (4.4)
Malignant neoplasm progression	3 (6.1)	0	3 (4.4)
Mucosal inflammation	1 (2.0)	2 (10.5)	3 (4.4)
Pulmonary embolism	3 (6.1)	0	3 (4.4)
Alanine aminotransferase increased	2 (4.1)	0	2 (2.9)
Atrial fibrillation	1 (2.0)	1 (5.3)	2 (2.9)
Colitis	2 (4.1)	0	2 (2.9)
Dyspnea	2 (4.1)	0	2 (2.9)
Fatigue	2 (4.1)	0	2 (2.9)
Hypoalbuminemia	2 (4.1)	0	2 (2.9)
Leukopenia	1 (2.0)	1 (5.3)	2 (2.9)
Malignant pleural effusion	1 (2.0)	1 (5.3)	2 (2.9)
Neutrophil count decreased	1 (2.0)	1 (5.3)	2 (2.9)
Pneumonia	2 (4.1)	0	2 (2.9)
Pyrexia	2 (4.1)	0	2 (2.9)
Rash	2 (4.1)	0	2 (2.9)
Respiratory failure	2 (4.1)	0	2 (2.9)
Thrombocytopenia	1 (2.0)	1 (5.3)	2 (2.9)

- The recommended phase 2 dose for the advanced solid tumor cohorts was relacorilant at 100 mg (continuous treatment) or 150 mg (intermittent treatment) with nab-pac 80 mg/m². Nab-pac exposure was similar to nab-pac 100 mg/m² monotherapy due to CYP3A4 inhibition by relacorilant.

EFFICACY

- Durable responses and disease control were observed. Best overall response (BOR) and disease control rate (DCR) >16 weeks in the response-evaluable population are shown in **Table 3**.
- Historical response rate is typically 0% in third-line PDAC and <15% in third-line ovarian cancer[9-11]

Table 3. Best overall response defined by RECIST v1.1 in response-evaluable patients

	Total (response evaluable)[a]		
	PDAC (n=25)	Ovarian cancer (n=11)	Other solid tumors (n=13)
CR, n	0	1	0
PR, n	4	2	3
SD, n	8	5	8
PD, n	13	3	2
DCR >16 wks, n/N (%) [range, wks]	7/25 (28.0) [17-52+]	5/11 (45.5) [18-65+]	7/13 (53.8) [17-80+]

CR, complete response; DCR, disease control rate; PDAC, pancreatic ductal adenocarcinoma; PR, partial response; PD, progressive disease; SD, stable disease; wks, weeks.
[a]Patients who received at least one dose of study drug and had at least one post-baseline tumor assessment.

Figure 2. Study treatment duration of patients with pancreatic ductal adenocarcinoma, ovarian cancer, and other solid tumors

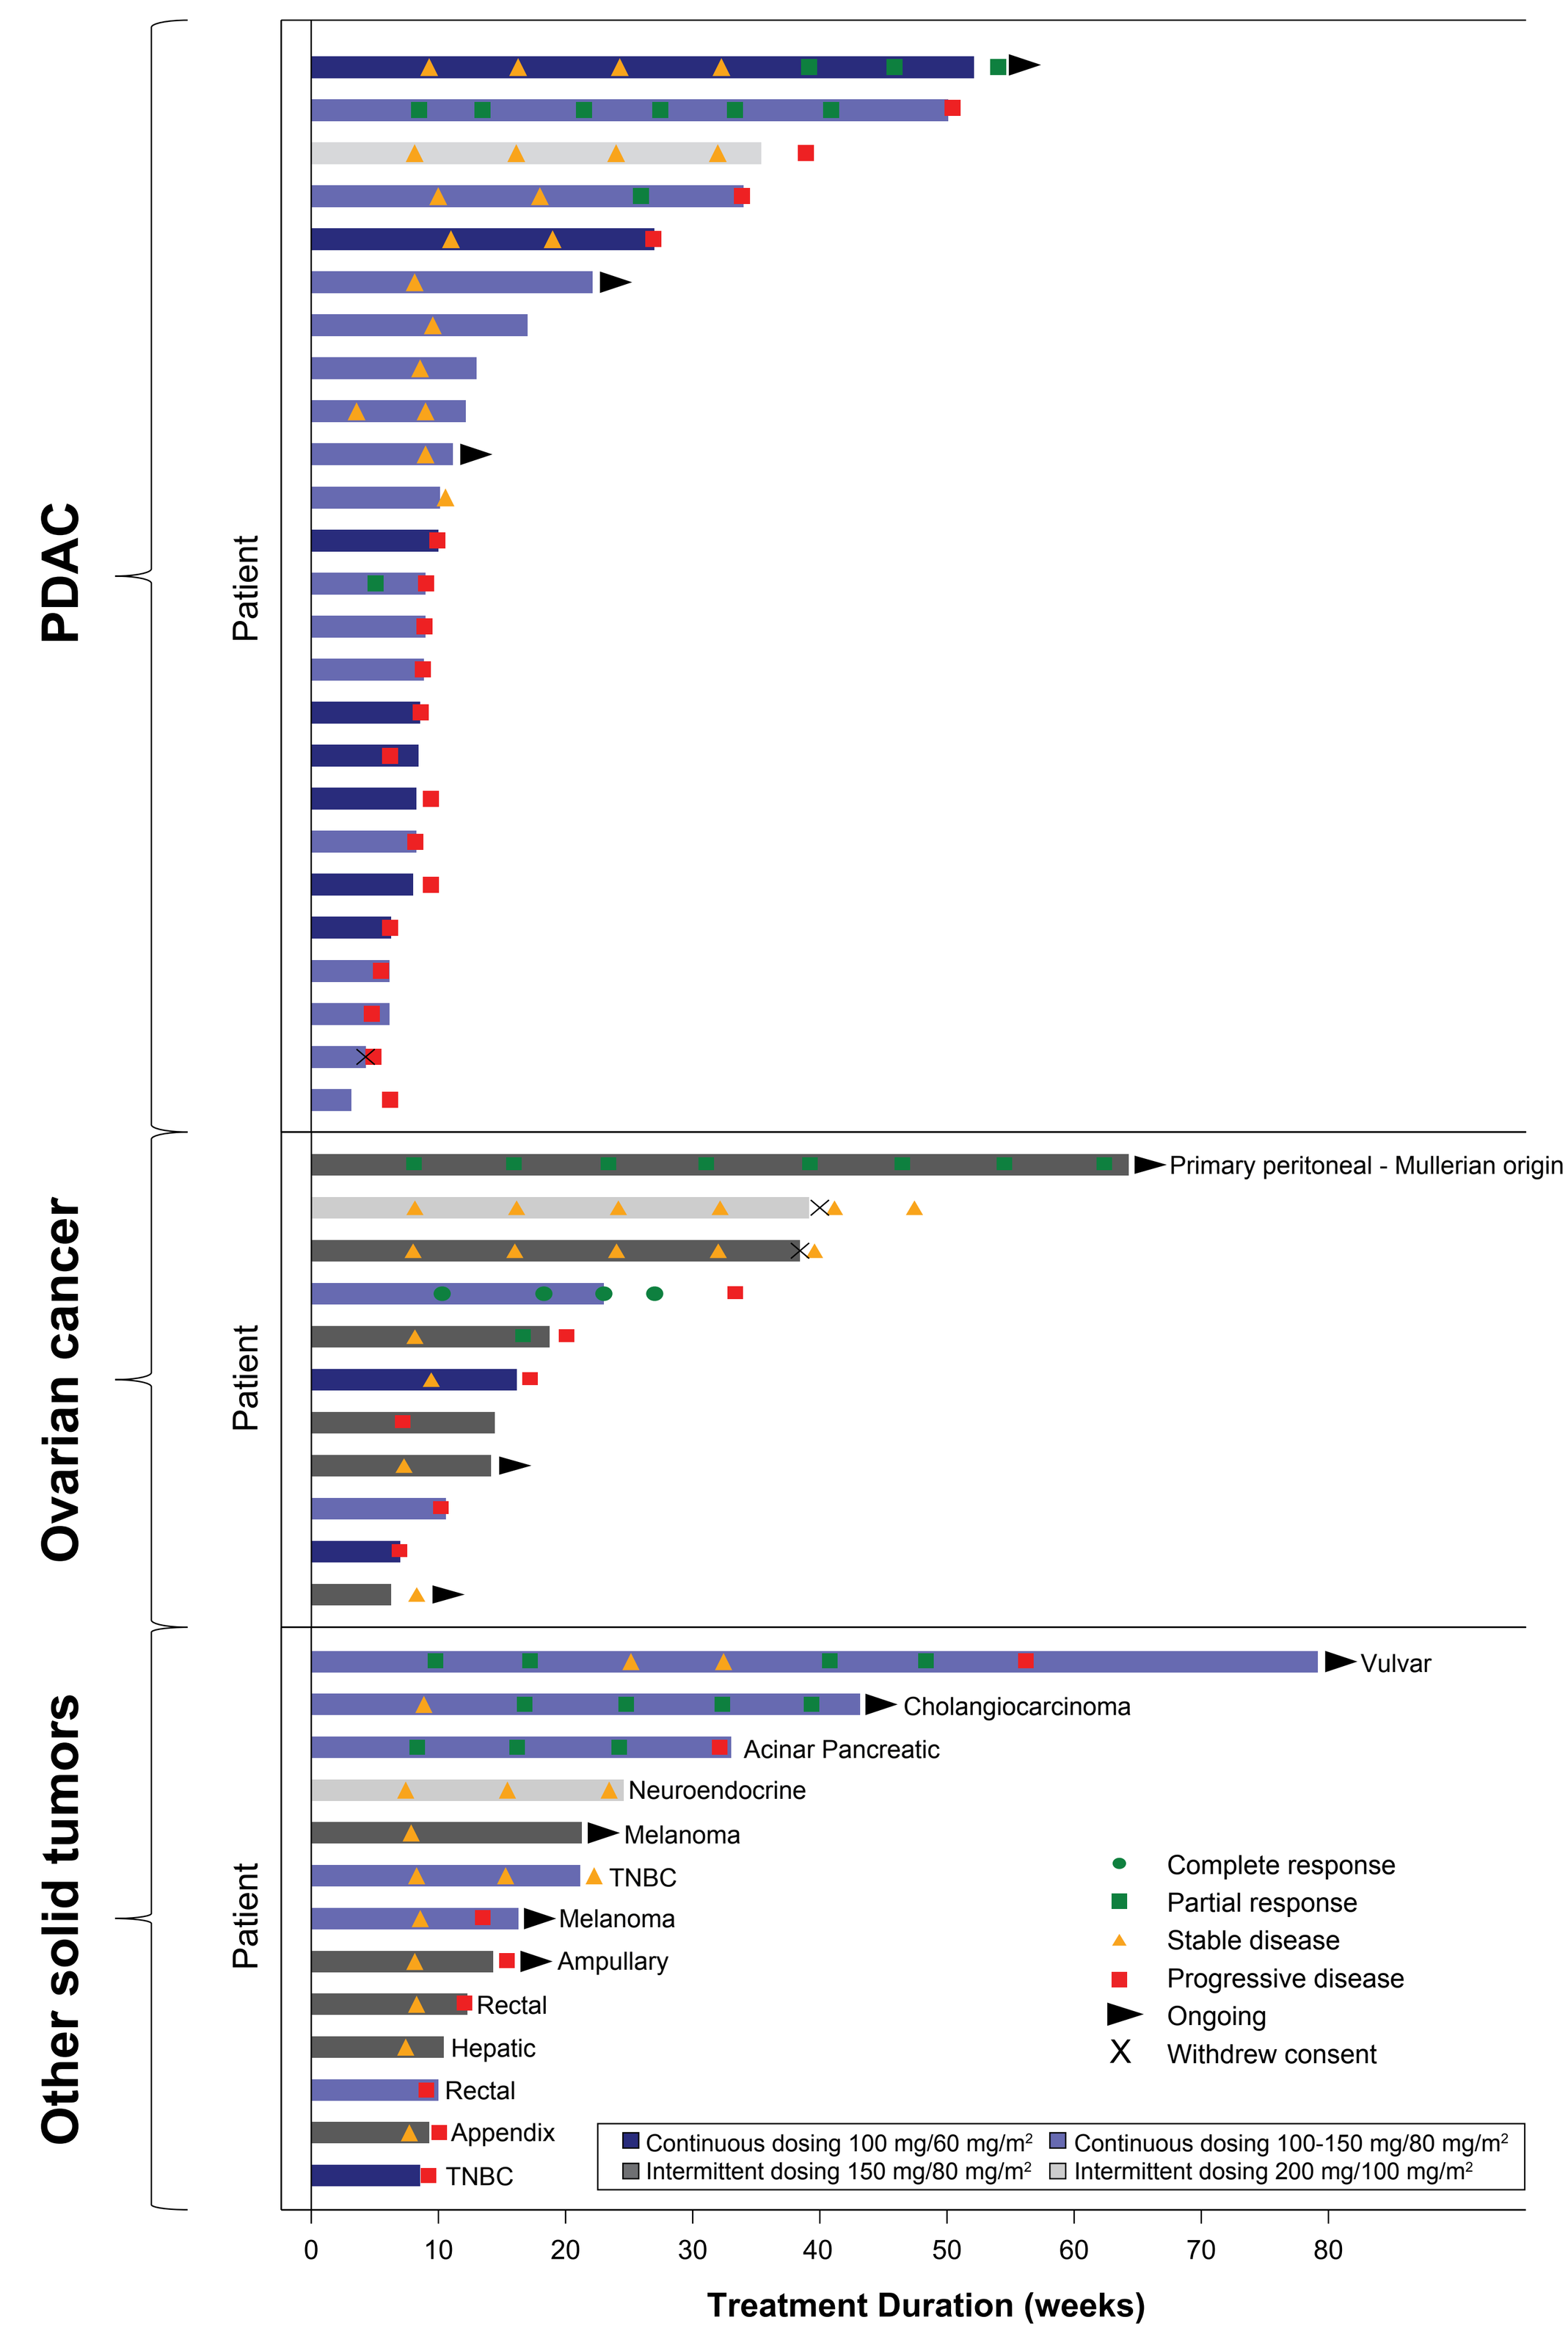

PDAC
Ovarian cancer
Other solid tumors
Patient
Primary peritoneal - Mullerian origin
Vulvar
Cholangiocarcinoma
Acinar Pancreatic
Neuroendocrine
Melanoma
TNBC
Melanoma
Ampullary
Rectal
Hepatic
Rectal
Appendix
TNBC
Complete response
Partial response
Stable disease
Progressive disease
Ongoing
Withdrew consent
Continuous dosing 100 mg/60 mg/m²
Continuous dosing 100-150 mg/80 mg/m²
Intermittent dosing 150 mg/80 mg/m²
Intermittent dosing 200 mg/100 mg/m²
Treatment Duration (weeks)

PDAC, pancreatic ductal adenocarcinoma; TNBC, triple negative breast cancer. Doses shown represent relacorilant/nab-pac dosing.

Figure 3. Longer duration of benefit achieved compared to prior duration of taxane treatment in patients with durable disease control (>16 weeks) and previously treated with a taxane

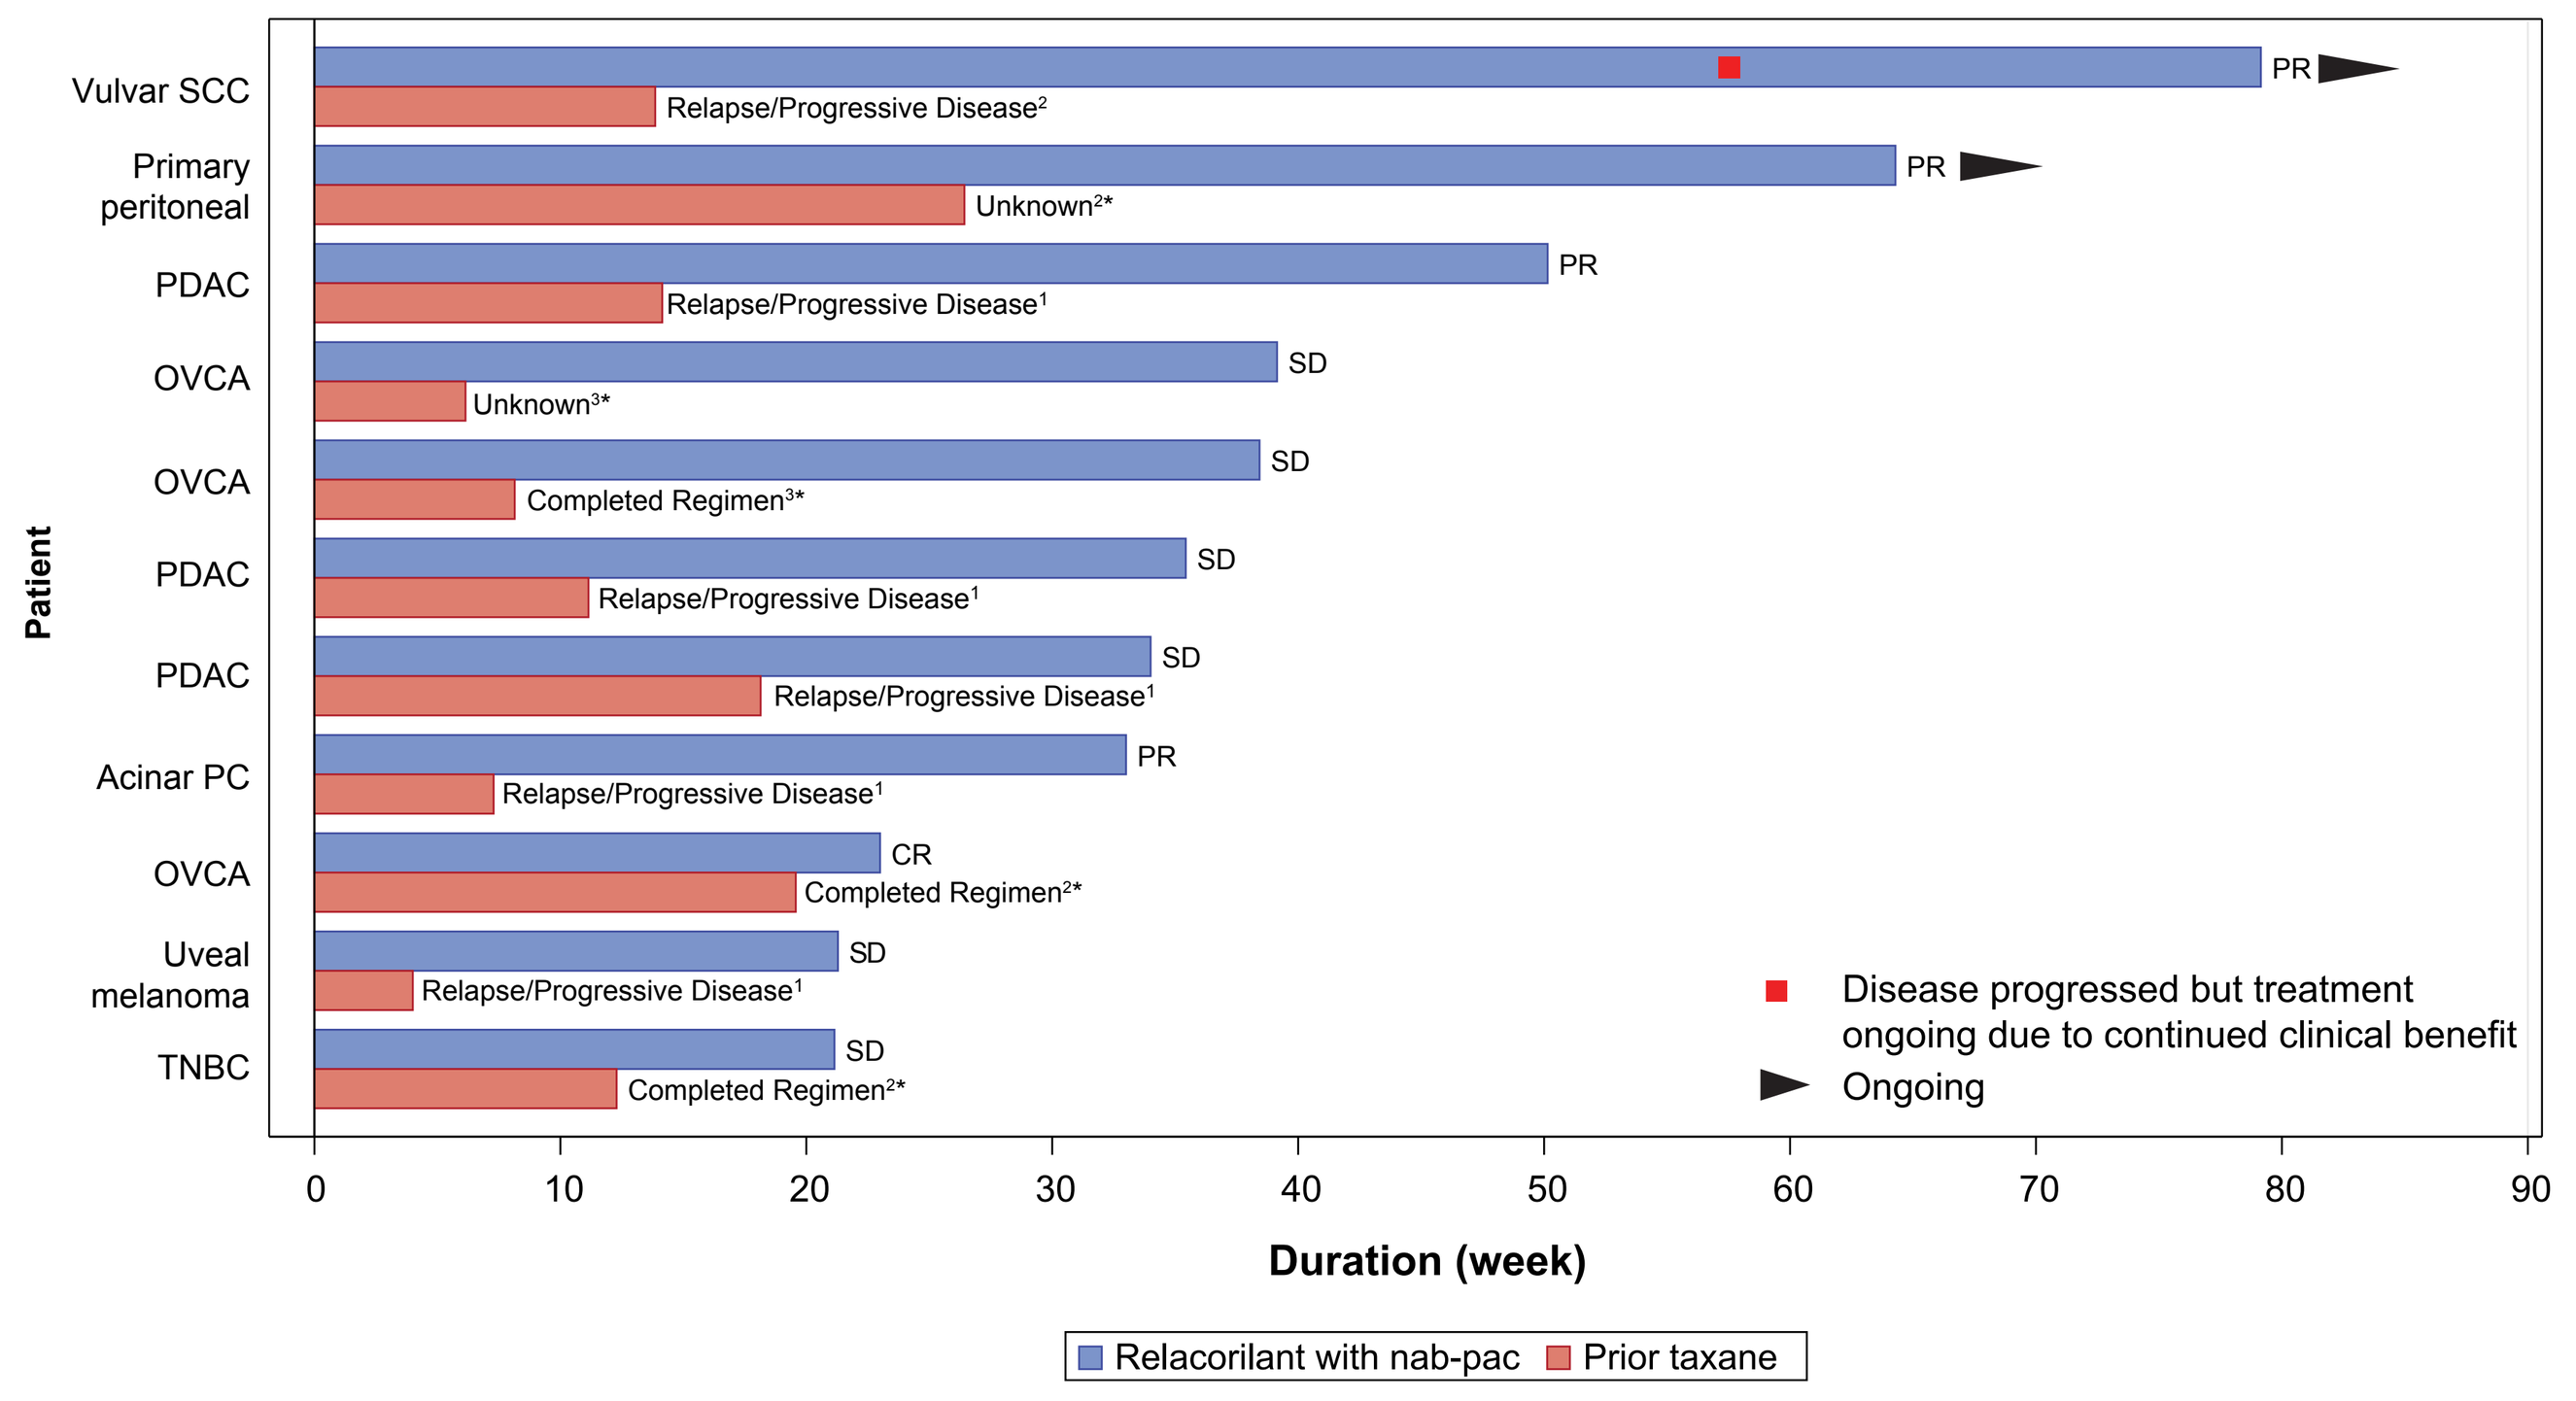

Vulvar SCC
Primary peritoneal
PDAC
OVCA
OVCA
PDAC
PDAC
Acinar PC
OVCA
Uveal melanoma
TNBC
Patient
PR
PR
PR
SD
SD
SD
SD
PR
CR
SD
SD
Relapse/Progressive Disease²
Unknown²*
Relapse/Progressive Disease¹
Unknown³*
Completed Regimen³*
Relapse/Progressive Disease¹
Relapse/Progressive Disease¹
Relapse/Progressive Disease¹
Completed Regimen²*
Relapse/Progressive Disease¹
Completed Regimen²*
Disease progressed but treatment ongoing due to continued clinical benefit
Ongoing
Duration (week)
Relacorilant with nab-pac
Prior taxane

[1]Nab-pac, [2]paclitaxel, [3]docetaxel.
*Adjuvant or neoadjuvant therapy.
CR, complete response; OVCA, ovarian cancer; PDAC, pancreatic ductal adenocarcinoma; PR, partial response; SCC, squamous cell carcinoma; SD, stable disease; TNBC, triple negative breast cancer.

- Third- and fourth-line pancreatic cancer: three patients achieved longer duration of benefit (2 PR, 1 SD) despite progression on prior taxane with time to progression on relacorilant + nab-pac that is 1.9-3.6x longer than prior taxane
- Ovarian cancer: all patients received prior taxane in the setting of adjuvant therapy. One patient received additional taxane in the metastatic setting, and this patient had time to progression on relacorilant + nab-pac that is 4.4x longer than prior taxane.
- Three additional PRs were observed: acinar pancreatic cancer, time to progression 32 wks (4.6x prior taxane); cholangiocarcinoma, disease control 40+ wks; vulvar SCC HPV+, time to progression 55 wks (3.9x prior taxane)

Figure 4. CT scans of (A) a patient with PDAC with metastasis to liver and (B) a patient with vulvar squamous cell carcinoma with metastasis to lung

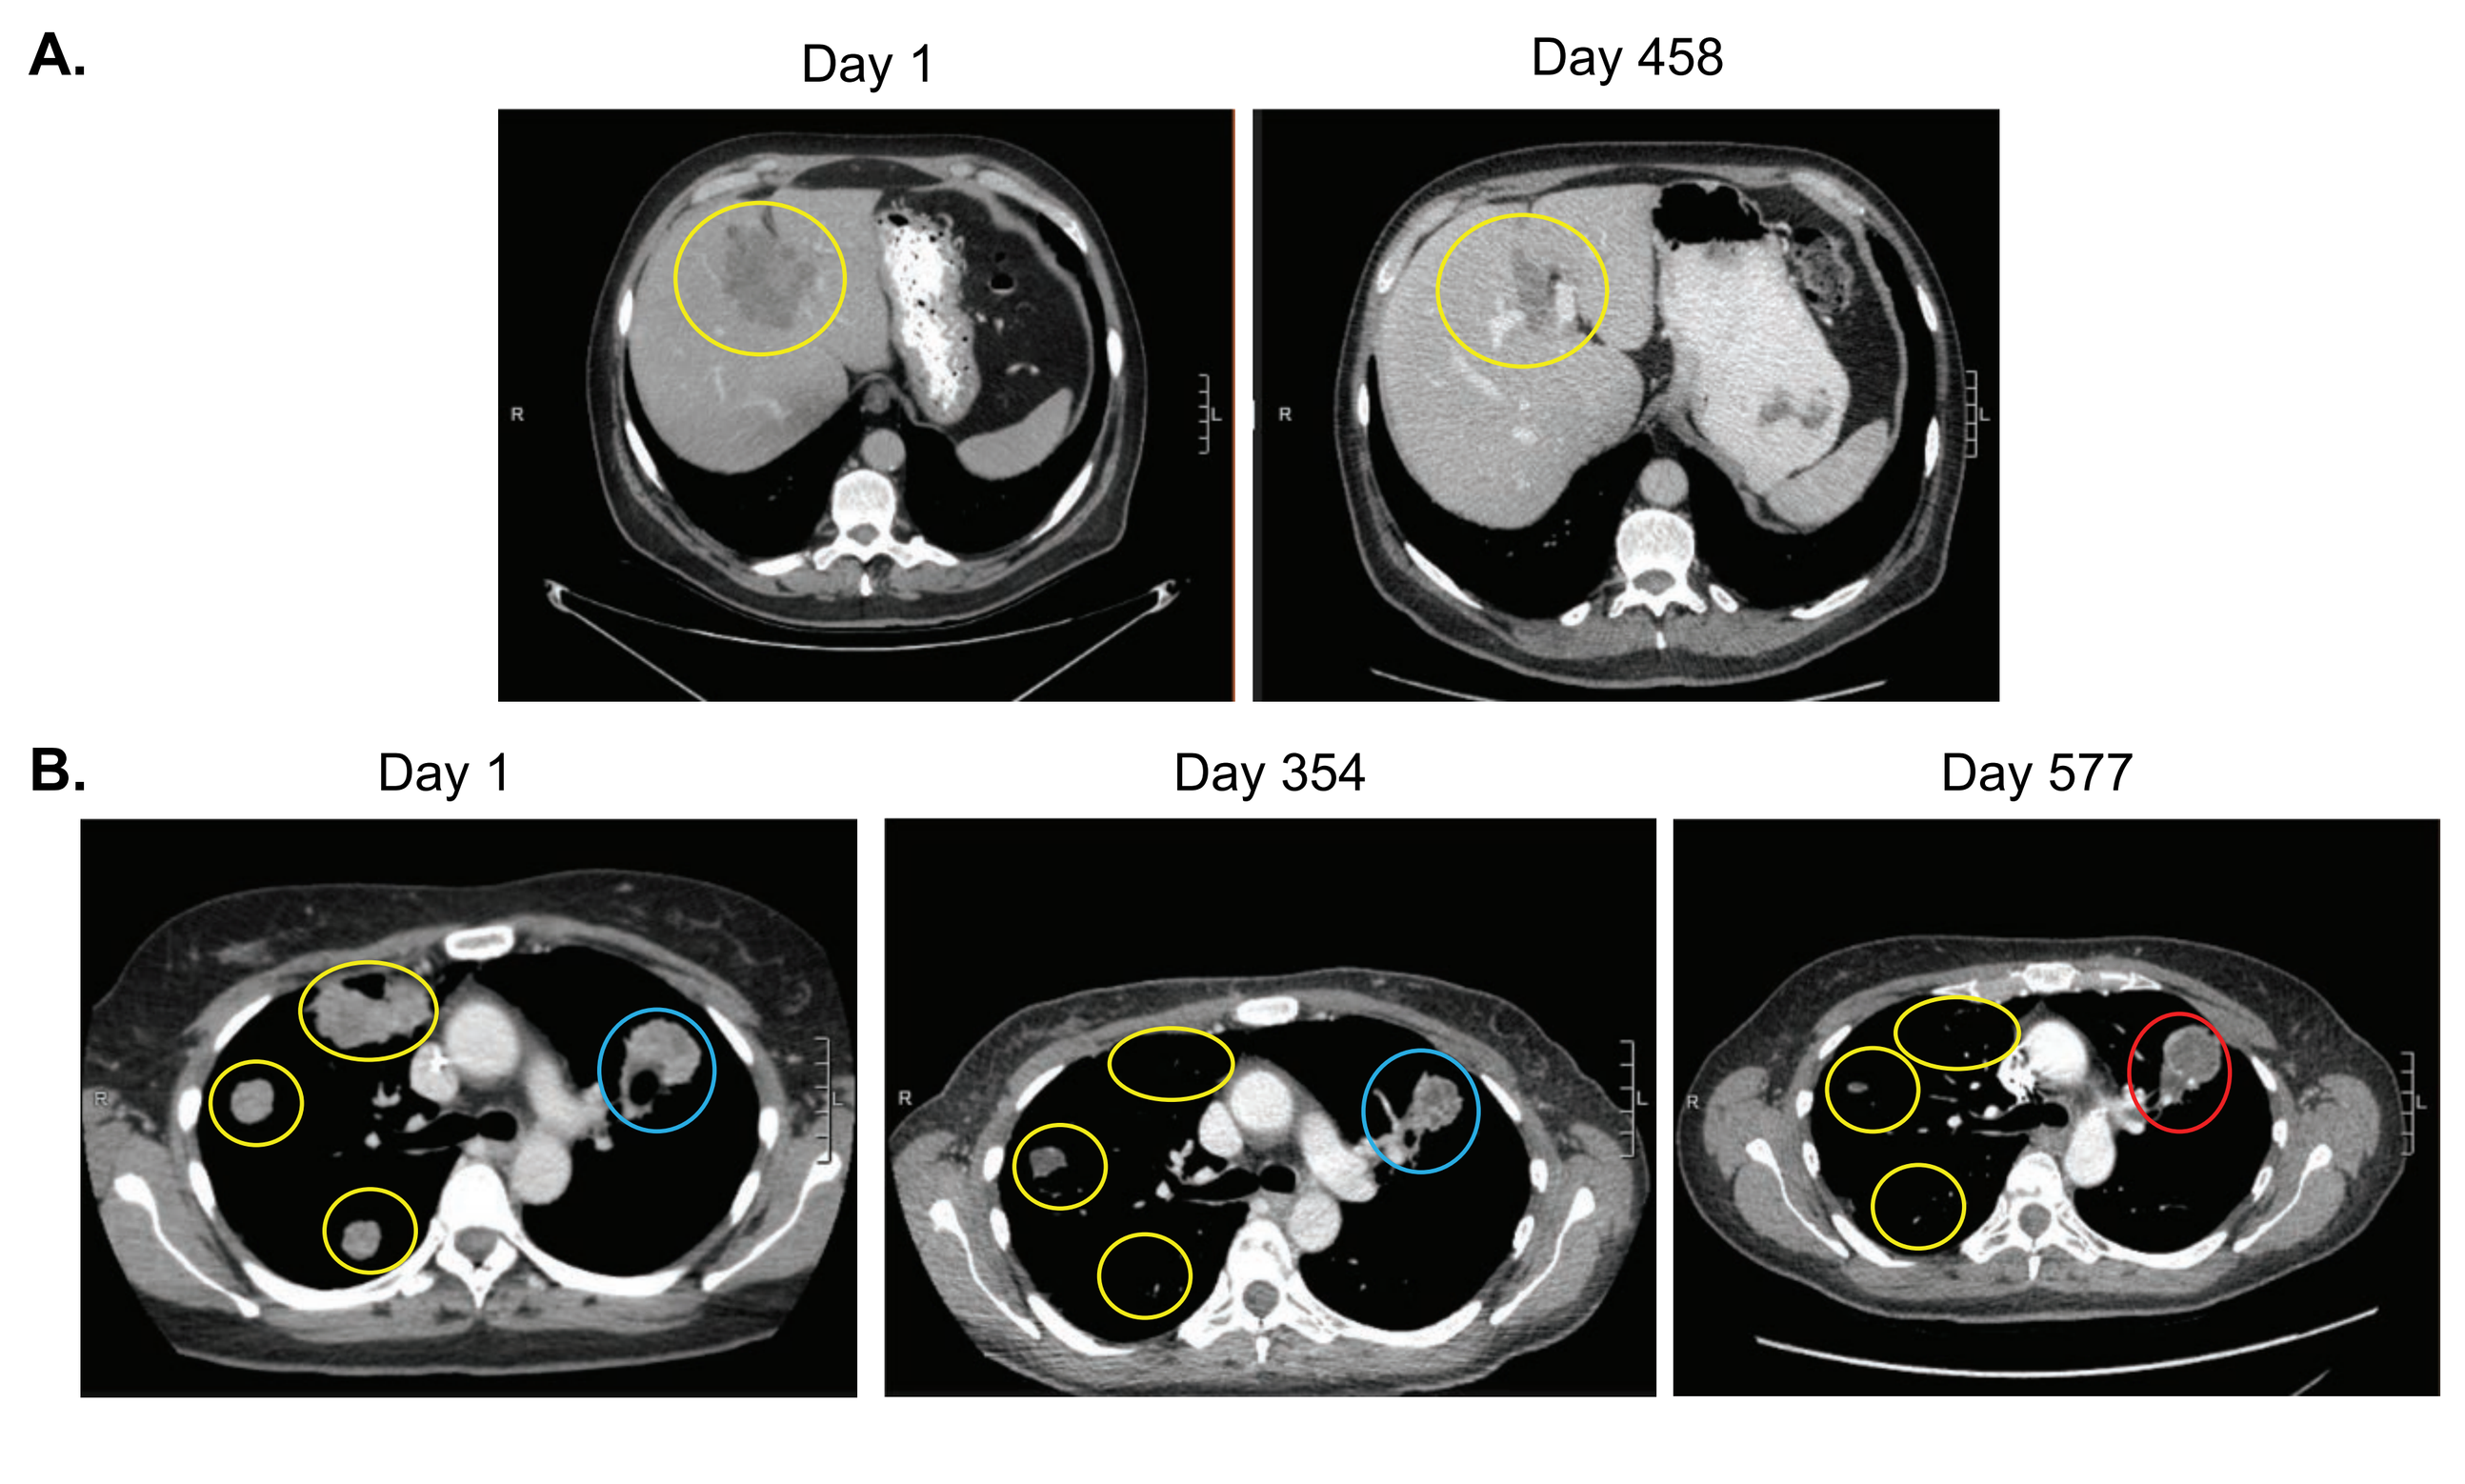

A.
Day 1
Day 458
B.
Day 1
Day 354
Day 577

(A) Liver lesion (yellow) size decreased from 8.0 cm to 4.9 cm, (B) Stable left lung lesion (blue) but significant reductions in sizes of right lung lesions (yellow). Radiotherapy was performed at Month 16 on symptomatic left lung lesion (red) with continued benefits seen in right lung lesions (yellow).

- GR immunohistochemistry (IHC) staining was positive (>10% of cells with 1+ or greater intensity) in all PDAC and ovarian cancer patients with available tumor sample for analysis. GR IHC staining was negative in two patients with triple negative breast cancer.

BIOMARKER: GR-REGULATED GENES WERE SUPPRESSED BY RELACORILANT + NAB-PAC

- GR-regulated genes are involved in inflammation, apoptosis, and immune function. Significantly greater number of GR-regulated genes were suppressed in patients with disease control vs. patients without disease control. Canonical GR-controlled genes such as COX2 and DUSP1 were suppressed in patients with disease control, but not in patients without disease control (**Figure 5**).
- GR-regulated gene changes did not correlate with levels of nab-pac exposure

Figure 5. Change in peripheral blood gene expression in patients receiving relacorilant + nab-pac

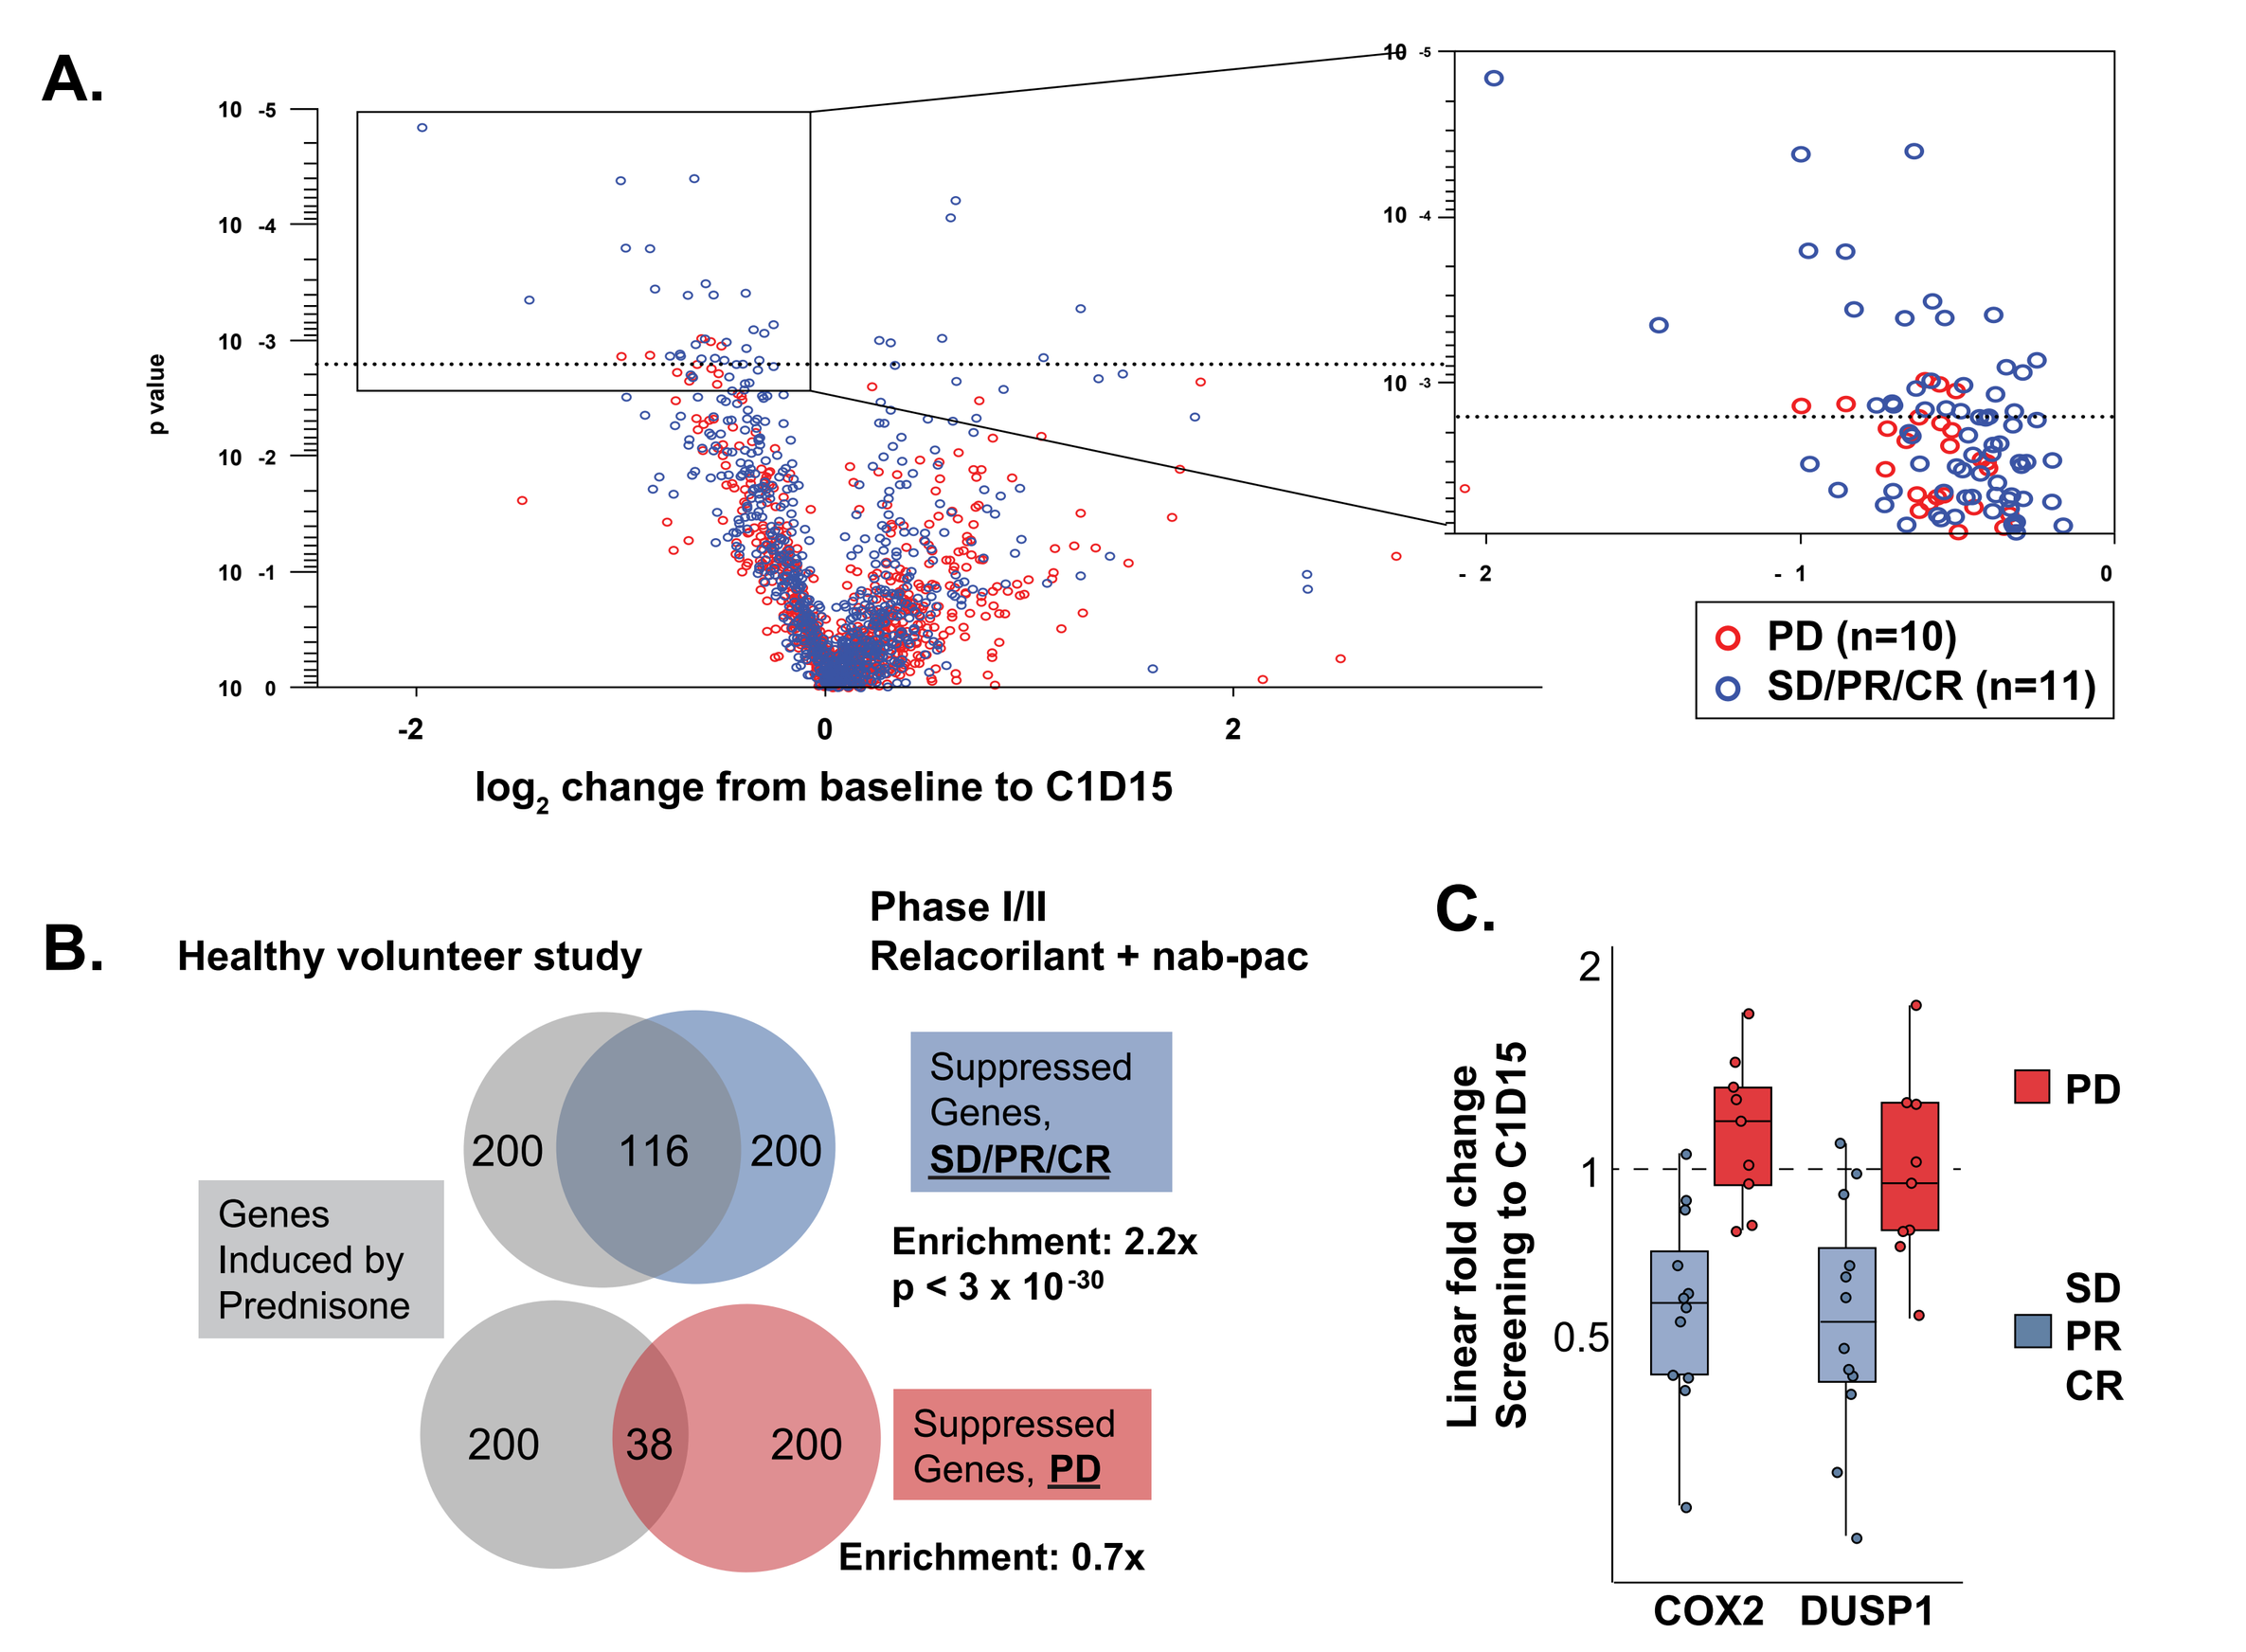

A.
p value
log₂ change from baseline to C1D15
PD (n=10)
SD/PR/CR (n=11)
B.
Healthy volunteer study
Phase I/II Relacorilant + nab-pac
Genes Induced by Prednisone
200
116
200
Suppressed Genes, SD/PR/CR
Enrichment: 2.2x
p < 3 x 10⁻³⁰
200
38
200
Suppressed Genes, PD
Enrichment: 0.7x
C.
Linear fold change Screening to C1D15
PD
SD
PR
CR
COX2
DUSP1

CR, complete response; GC, glucocorticoid; GR, glucocorticoid receptor, PD, progressive disease; PR, partial response; SD, stable disease.
(A) Effect of relacorilant + nab-pac on transcription of C1D15 versus pre-treatment in whole blood from patients with SD, PR, or CR vs PD; transcriptional changes were separated by best overall response. Inset details genes downregulated in SD/PR/CR (92 genes) vs PD (30 genes) patients. Dotted line represents an adjusted p-value of 0.05. (B) Top 200 genes with the largest change following 4 hours following treatment with 25 mg prednisone in a separate study of healthy subjects. GC-regulated genes were suppressed by relacorilant + nab-pac. A significant overlap in genes induced by prednisone in healthy subjects and suppressed in this study in patients receiving relacorilant + nab-pac was observed only in the patients with a best overall response of SD, PR, or CR. (C) Canonical GR-controlled genes COX2 and DUSP1 are suppressed in patients with SD, PR, or CR but not patients with PD. Fold changes (1 represents no change) were significantly different between SD, PR, and CR patients vs PD patients for both COX2 (unadjusted p=0.00015) and DUSP1 (unadjusted p=0.0055).

CONCLUSIONS

- Relacorilant demonstrated clinical activity in patients with metastatic PDAC, ovarian cancer, and other solid tumors by enhancing nab-pac activity and suppressing GC-controlled genes
 - Time to progression was often several-fold longer than previously achieved on taxane therapy
 - GR-specific pharmacodynamic responses were observed in patients with disease control, underscoring the contribution of relacorilant to disease control
- Observed adverse events reflected the advanced disease and treatment history of the patients. Neutropenia was manageable with prophylactic granulocyte colony-stimulating factor.
- Further evaluation in ovarian cancer (NCT03776812), metastatic PDAC, and other tumors is ongoing

REFERENCES

1. Baker GM, et al. *Cancer Manag Res.* 2015;7:361-8.
2. Block TS, et al. *Cancer Manag Res.* 2017;9:65-72.
3. Liu L, et al. *Cell Death Dis.* 2017;8(10):e3064.
4. Veneris JT, et al. *Gynecol Oncol.* 2017;146(1):153-60.
5. Zhang C, et al. *Int J Oncol.* 2006;28(2):551-8.
6. Stringer-Reasor EM, et al. *Gynecol Oncol.* 2015;138(3):656-62.
7. Zhang C, et al. *BMC Cancer.* 2006;6:61.
8. Hunt HJ, et al. *J Med Chem.* 2017;60(8):3405-21.
9. Peddi, et al. J Gastrointest Oncol. 2013;4370-3.
10. Macarulla, et al. *J of Clinical Oncology.* 2017;35:4127.
11. Bruchim, et al. *Eur J Obstet Gynecol Reprod Biol.* 2013;166:94-8.

ACKNOWLEDGEMENTS

This study was funded by Corcept Therapeutics. Funding for design and production support was provided by Corcept Therapeutics to MedVal Scientific Information Services, LLC. Medical writing assistance was provided by Meghan Sullivan, PhD, of MedVal Scientific Information Services (Princeton, NJ, USA), and funded by Corcept Therapeutics. The authors thank Radhika Sharma for project management, Eric Fritz for data management, and Cristina Tudor and Peiling Ho for statistical analysis.

Clinicaltrials.gov: NCT02762981

Presented at the American Society of Clinical Oncology Annual Meeting, May 31 – June 4, 2019, Chicago, IL