LB-130

Suppression of Tumor Immune Activity in Adrenocortical Carcinoma with Excess Glucocorticoid

Andrew E. Greenstein[1], Mouhammed Amir Habra[2], Marzena Mura[3], Paweł Biernat[3], Marek J. Piatek[3], Andreas Grauer[1], and Stacie Peacock Shepherd[5]
[1]Corcept Therapeutics, Menlo Park, CA, USA; [2]MD Anderson Cancer Center, Houston, TX, USA; [3]Ardigen SA, Krakow, Poland; [5]Formerly Corcept Therapeutics, Menlo Park, CA, USA
Contact: agreenstein@corcept.com

BACKGROUND

- Elevated glucocorticoid (GC) activity has been implicated in the pathophysiology of multiple cancer types, including breast, ovarian, squamous cell, and cervical cancer, as well as lymphomas [1-5].
 - Patients administered GC prior to immune checkpoint inhibitor (ICI) therapy are reported to experience worse outcomes across multiple oncology indications [6-8].
- Adrenocortical carcinoma (ACC), a rare endocrine cancer, is often accompanied by secretion of steroid hormones.
 - Approximately half of ACC tumors are functional, with 43% producing GC, either alone or in combination with other hormones [9].
 - Therapies to reduce tumor burden for patients with ACC are limited and treatment outcomes are generally poor.
 - Presence of GC excess (GC+) is correlated with decreased overall survival and disease-free survival in patients with ACC [9].
 - Furthermore, ACC patients with GC+ tumors show poor responses to ICI therapy [10, 11].
- We hypothesize that the broad immunosuppressive effects of GC may limit tumor immune response and ICI efficacy, which may be improved by glucocorticoid receptor (GR) antagonism.
- General hormone and GC abundance are annotated in The Cancer Genome Atlas (TCGA) ACC dataset [12], which provides a unique test case to assess correlates of GC activity.

▶ Here, we present an ACC multi-omics analysis to identify the molecular consequences of GC activity and assess the rationale for combining relacorilant (CORT125134, Corcept Therapeutics), an investigational GR antagonist, with an ICI in the treatment of ACC with GC excess.

METHODS

- GC or other hormone excess (based on clinical signs and symptoms or biochemical evidence), mRNA expression, DNA mutation, and DNA methylation data from distinct adrenal resections (n=71) were accessed via the TCGA (www.cancer.gov/tcga). Two sarcomatoid cases were excluded from the analysis.
- 394,036 methylation probes were analyzed and data were normalized using beta-mixture quantile normalization.
- To deconvolute immune cell type abundance, xCell was applied to the mRNA data [13].
- Tumor cases were scored using a published GR activity signature [14].
- Random forests were used to derive a gene signature predictive of GC+ tumors.
 - Signature genes were identified by bootstrapping random forests on random subsets comprising 80% of the data and comparing the mean bootstrapped importance of genes with a threshold value.
 - The threshold value was calculated by applying the same procedure to a random forest predicting randomized labels instead of the true GC+/- labels to simulate lack of signal. The 99.9th quantile of gene importance was selected as the threshold.

RESULTS

CLASSIFYING ACC USING GC STATUS

- Using mRNA data in TCGA, genes that differed significantly by general hormone or GC status (>2-fold change and adjusted P≤.05) were identified.
- Presence or absence of GC excess (GC+/-) was identified as affecting the largest number of genes (858 genes, Comparison 1 in **Figure 1** and colored regions in **Figure 2**).
 - Presence vs absence of any hormone (H+/-) led to a significant difference in 439 genes (Comparison 2).
 - There was no significant difference between H- tumors and those expressing only non-GC hormones (NGC+, Comparison 3).
 - A comparison of NGC+ vs GC+ revealed 185 significantly different genes (Comparison 4).
- Kyoto Encyclopedia of Genes and Genomes (KEGG) pathway analysis identified higher gene expression in GC+ cases in several steroid-synthesis pathways and lower expression in a number of immune-related pathways (**Figure 3**).

Figure 1. Classification of TCGA ACC Tumors (n=71) by Hormone Status. Among the 4 comparisons performed, presence or absence of GC excess (GC+/-) was associated with the largest number of significantly different genes in ACC.

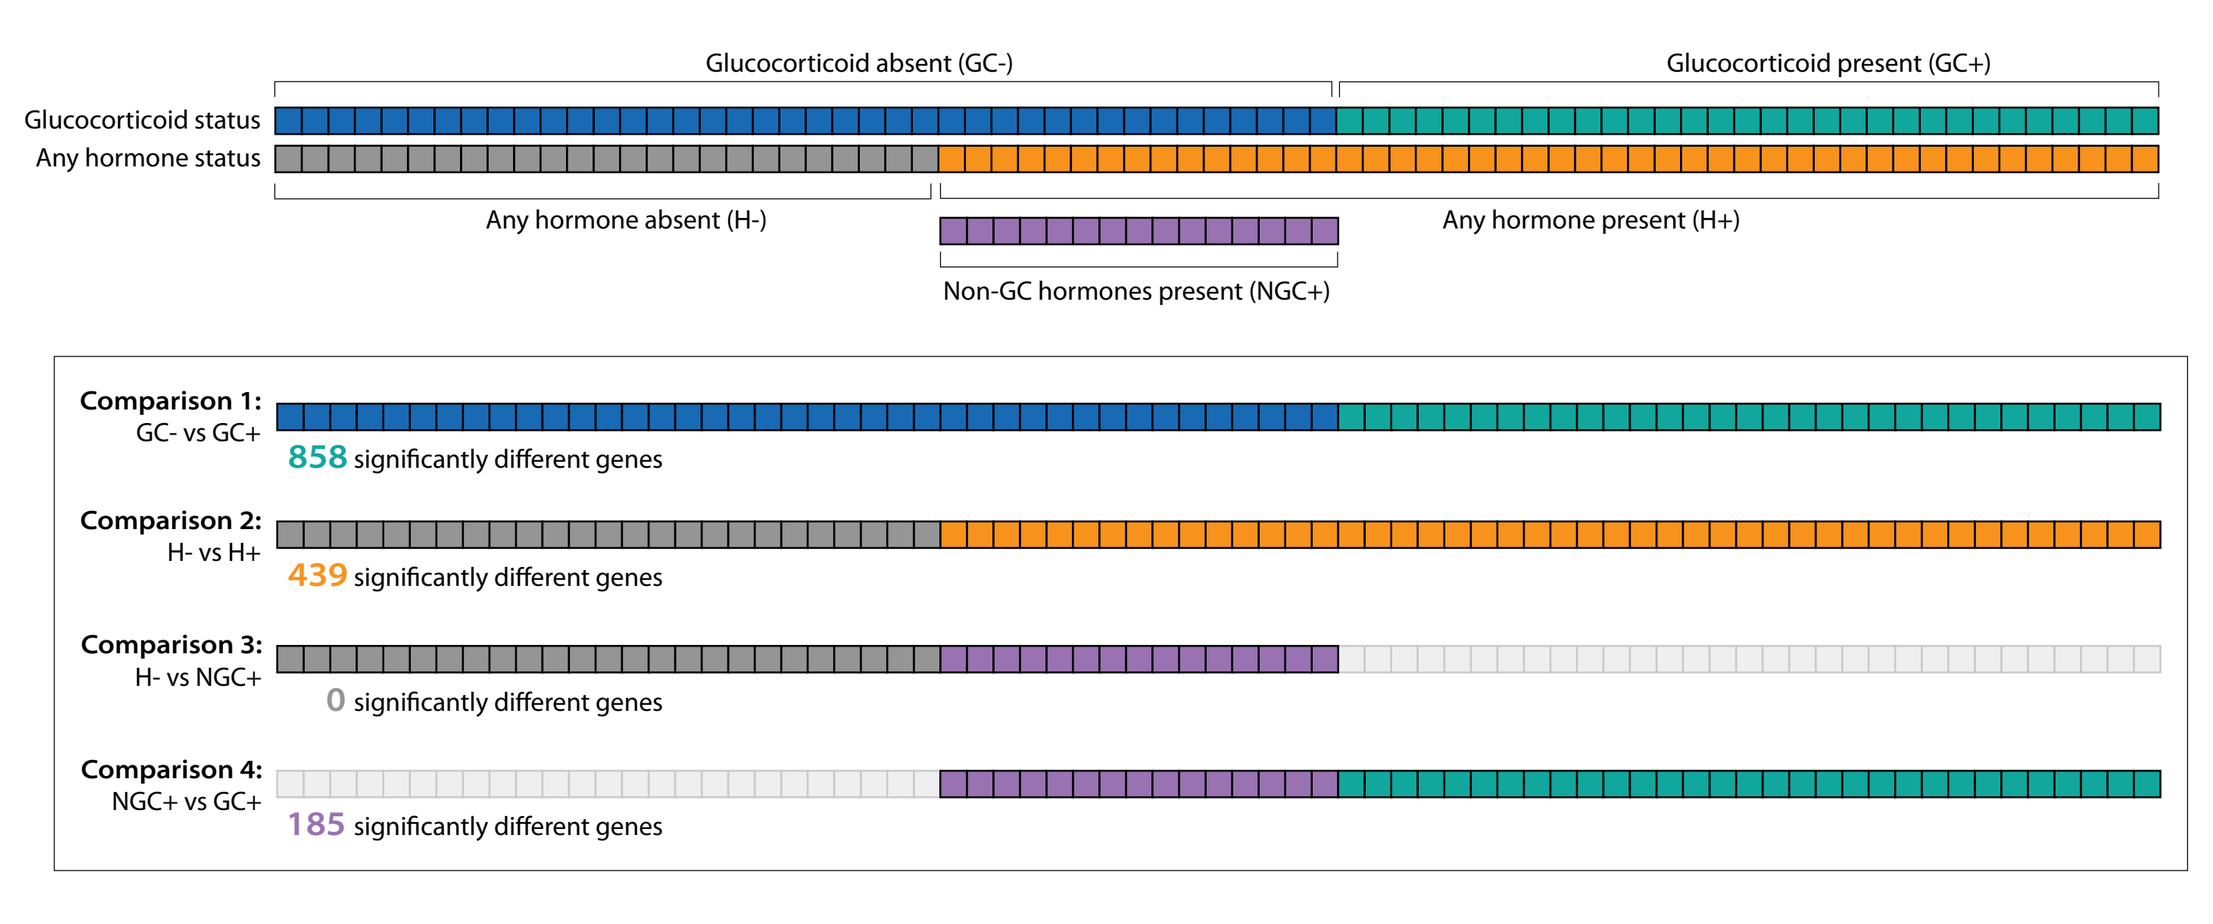
Glucocorticoid absent (GC-)
Glucocorticoid present (GC+)
Glucocorticoid status
Any hormone status
Any hormone absent (H-)
Any hormone present (H+)
Non-GC hormones present (NGC+)
Comparison 1: GC- vs GC+
858 significantly different genes
Comparison 2: H- vs H+
439 significantly different genes
Comparison 3: H- vs NGC+
0 significantly different genes
Comparison 4: NGC+ vs GC+
185 significantly different genes

Figure 2. Transcriptional Effects of GC Excess. Expression of 858 genes was found to be significantly affected by GC excess. Genes with higher expression in GC+ cases (*P*≤.05 and >2-fold change in expression compared to GC-) are shown on the right (red). Those with lower expression in GC+ cases are shown on the left (blue).

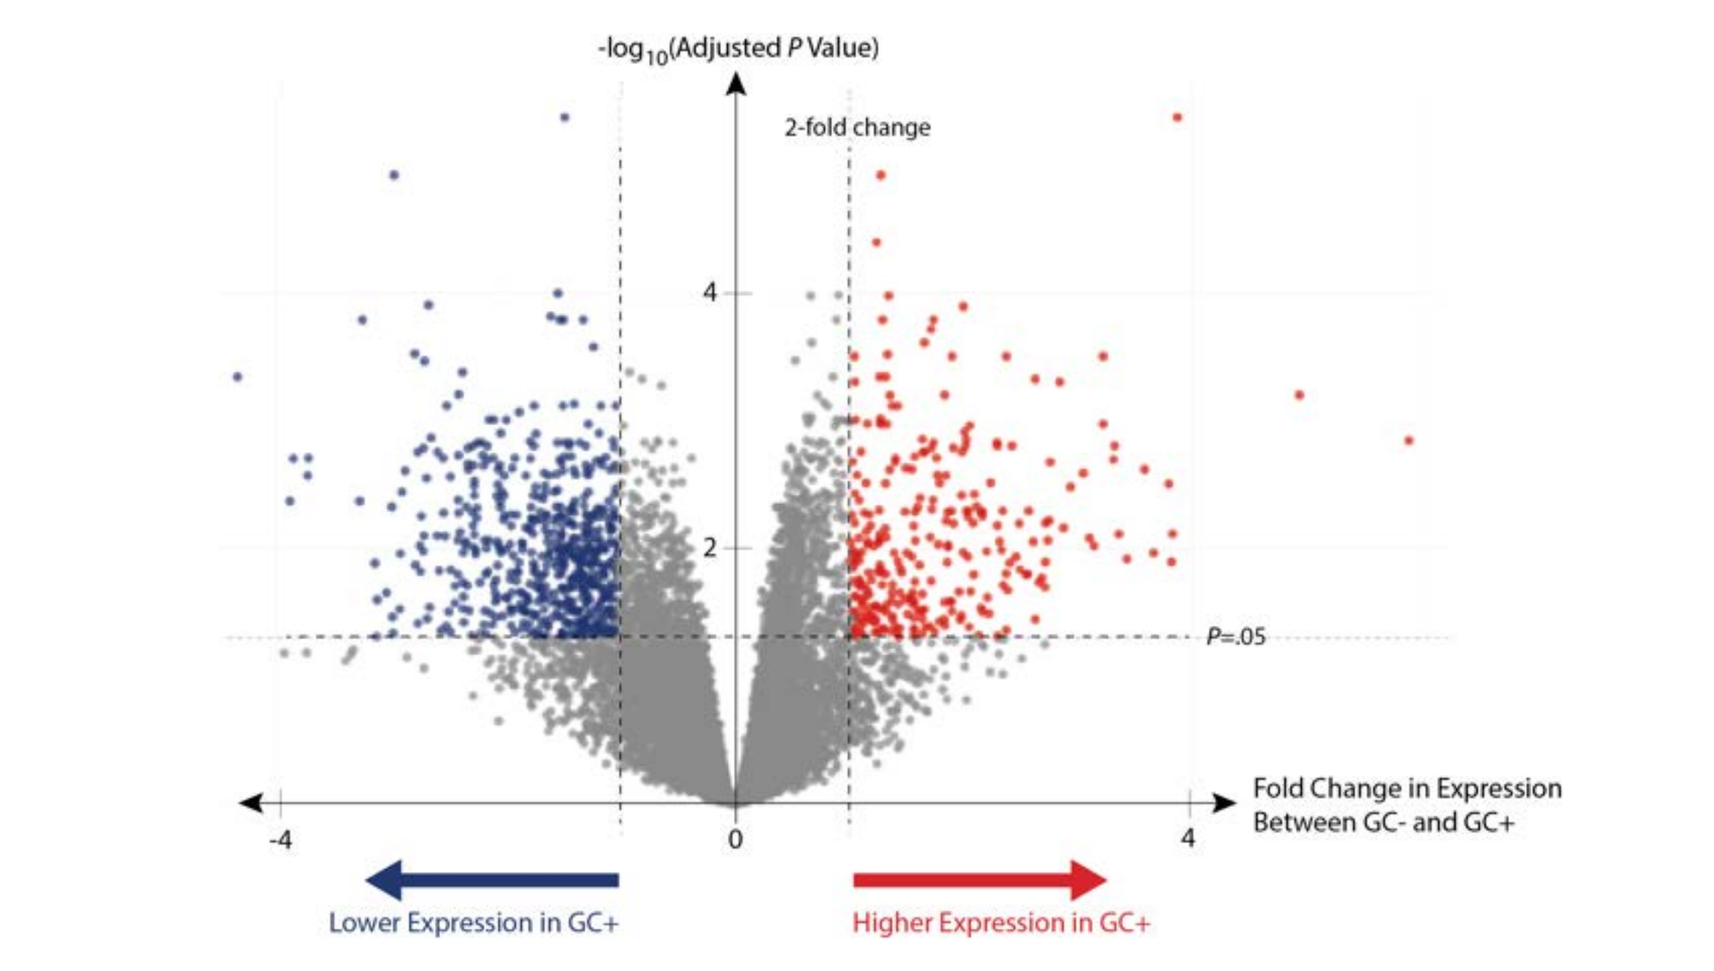
-log10(Adjusted P Value)
2-fold change
P=.05
Fold Change in Expression Between GC- and GC+
Lower Expression in GC+
Higher Expression in GC+

Figure 3. Overrepresented Downregulated and Upregulated Pathways in GC+ ACC Cases. Genes affected by GC excess include immune and steroid synthesis processes.

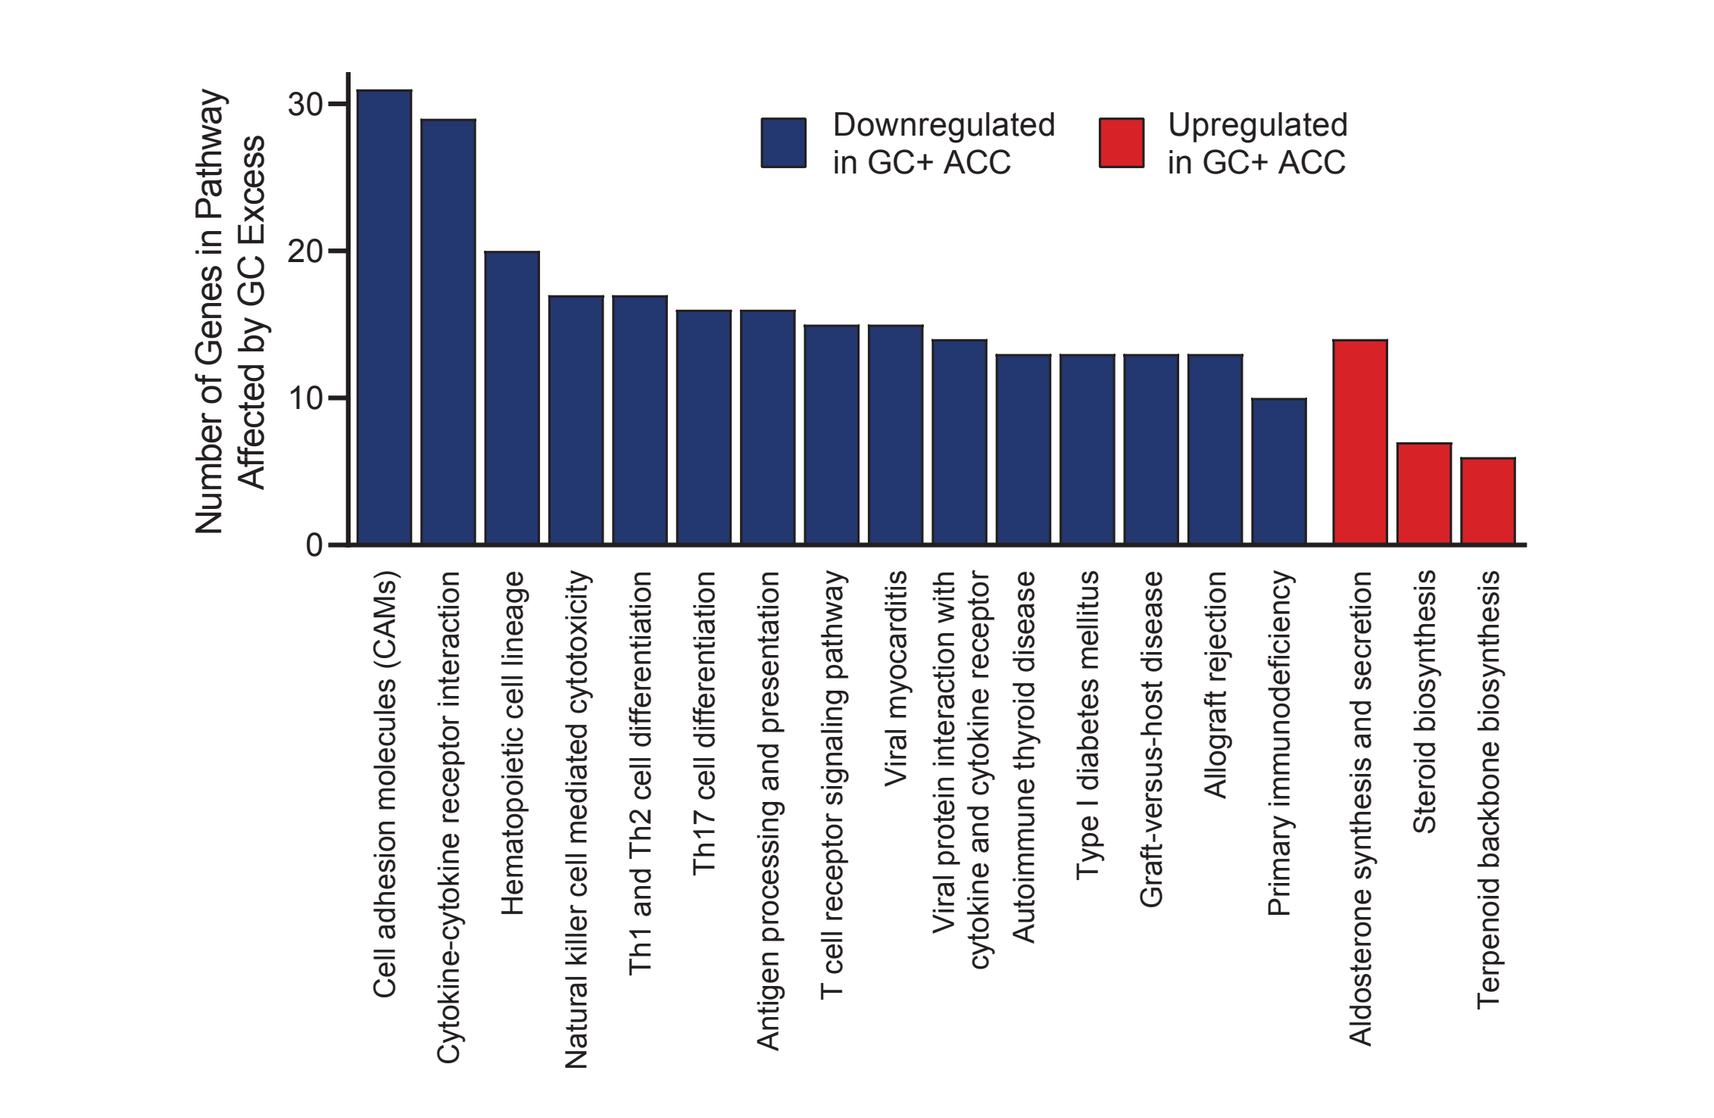
Number of Genes in Pathway Affected by GC Excess
Downregulated in GC+ ACC
Upregulated in GC+ ACC
Cell adhesion molecules (CAMs)
Cytokine-cytokine receptor interaction
Hematopoietic cell lineage
Natural killer cell mediated cytotoxicity
Th1 and Th2 cell differentiation
Th17 cell differentiation
Antigen processing and presentation
T cell receptor signaling pathway
Viral myocarditis
Viral protein interaction with cytokine and cytokine receptor
Autoimmune thyroid disease
Type I diabetes mellitus
Graft-versus-host disease
Allograft rejection
Primary immunodeficiency
Aldosterone synthesis and secretion
Steroid biosynthesis
Terpenoid backbone biosynthesis

IMMUNE GENE SUPPRESSION IS ASSOCIATED WITH GC PRODUCTION

- Unsupervised clustering of normalized gene expression for the T-cell receptor signaling and natural-killer-cell-mediated cytotoxicity KEGG pathways showed lower gene expression in GC+ cases (**Figure 4**).
- When clustering GC+ and GC- separately, GC+ cases trended toward lower expression in these two immune-related pathways (**Figure 5**).

Figure 4. Unsupervised Clustering of Normalized Gene Expression for 2 Immune KEGG Pathways. Pathways shown include T-cell receptor signaling and natural-killer-cell-mediated cytotoxicity. The top 2 rows indicate GC and general hormone status for each tumor (black: GC+/H+, white: GC-/H-), shades of blue/red show normalized gene expression for each tumor, with darker blue corresponding to lower expression. When clustering by gene expression, GC+ cases appear toward the right of the figure, where many genes show lower expression.

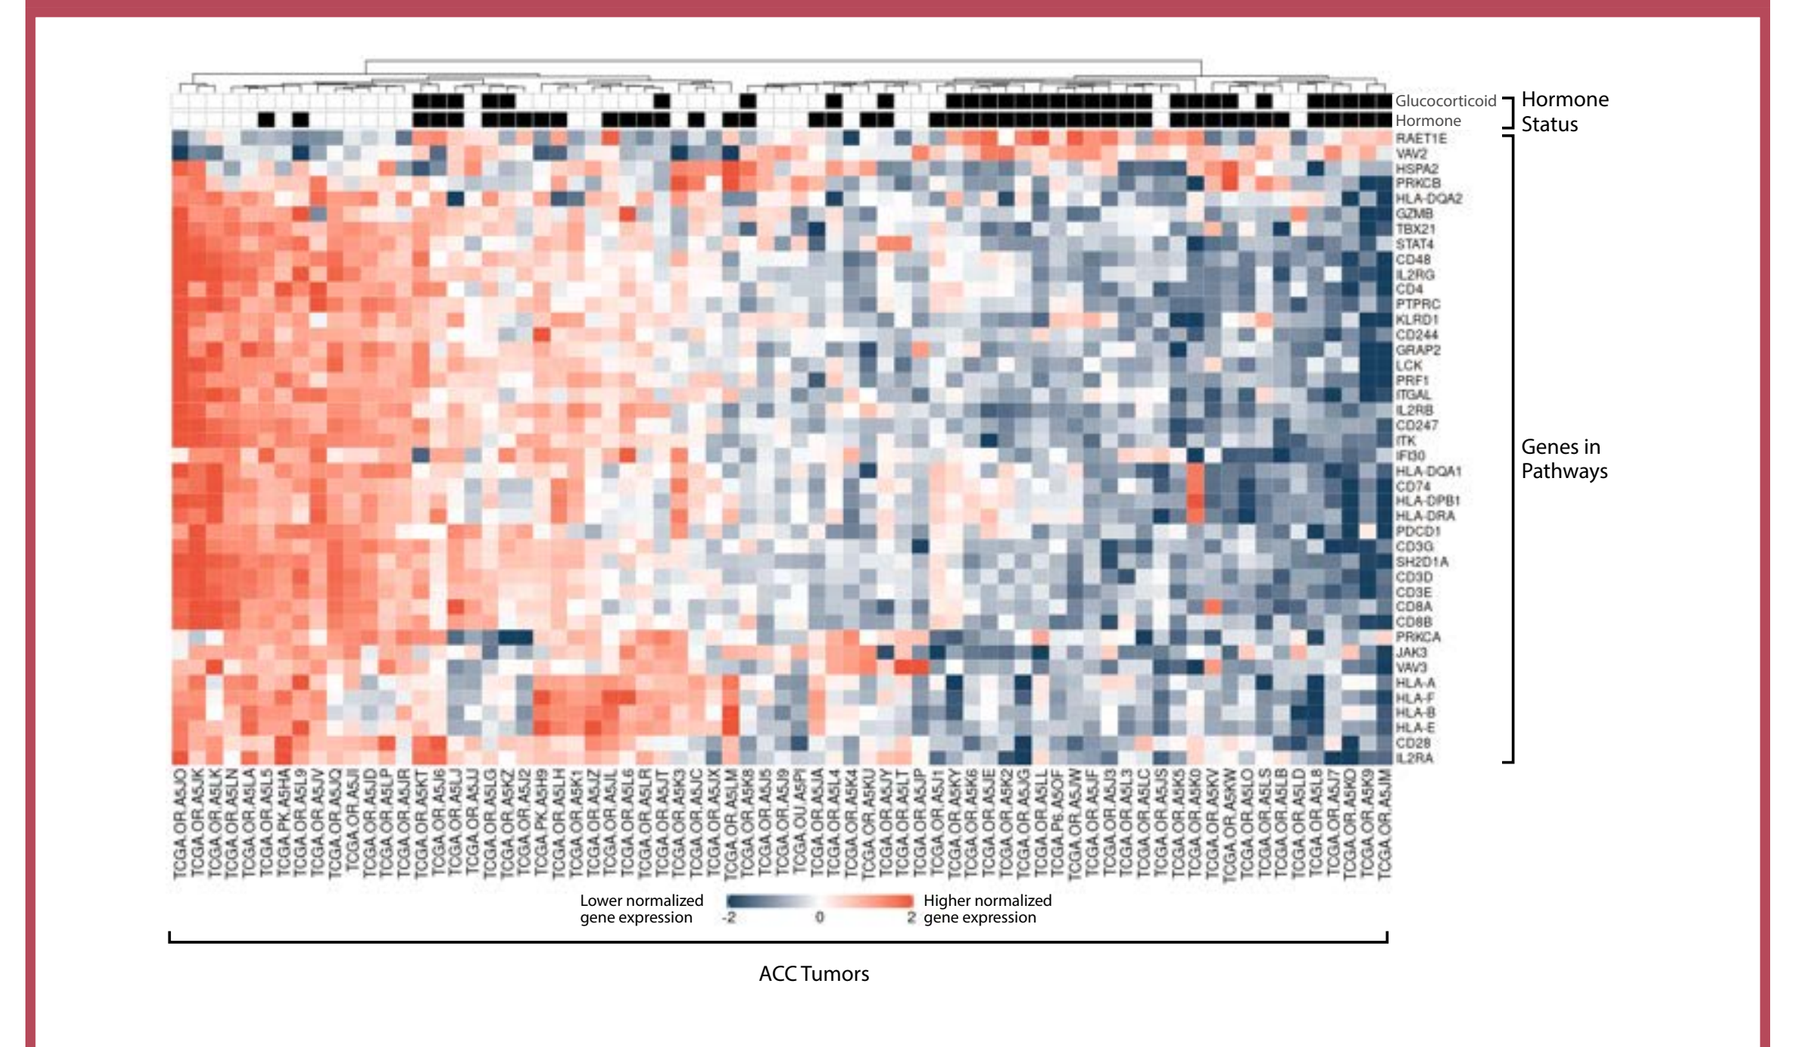
Hormone Status
Genes in Pathways
Lower normalized gene expression
Higher normalized gene expression
ACC Tumors

Figure 5. Supervised Clustering of Normalized Gene Expression for the 2 Immune KEGG Pathways Shown in Figure 4. In GC+ cases (cluster on the right), lower expression of genes dominates these pathways (darker blue).

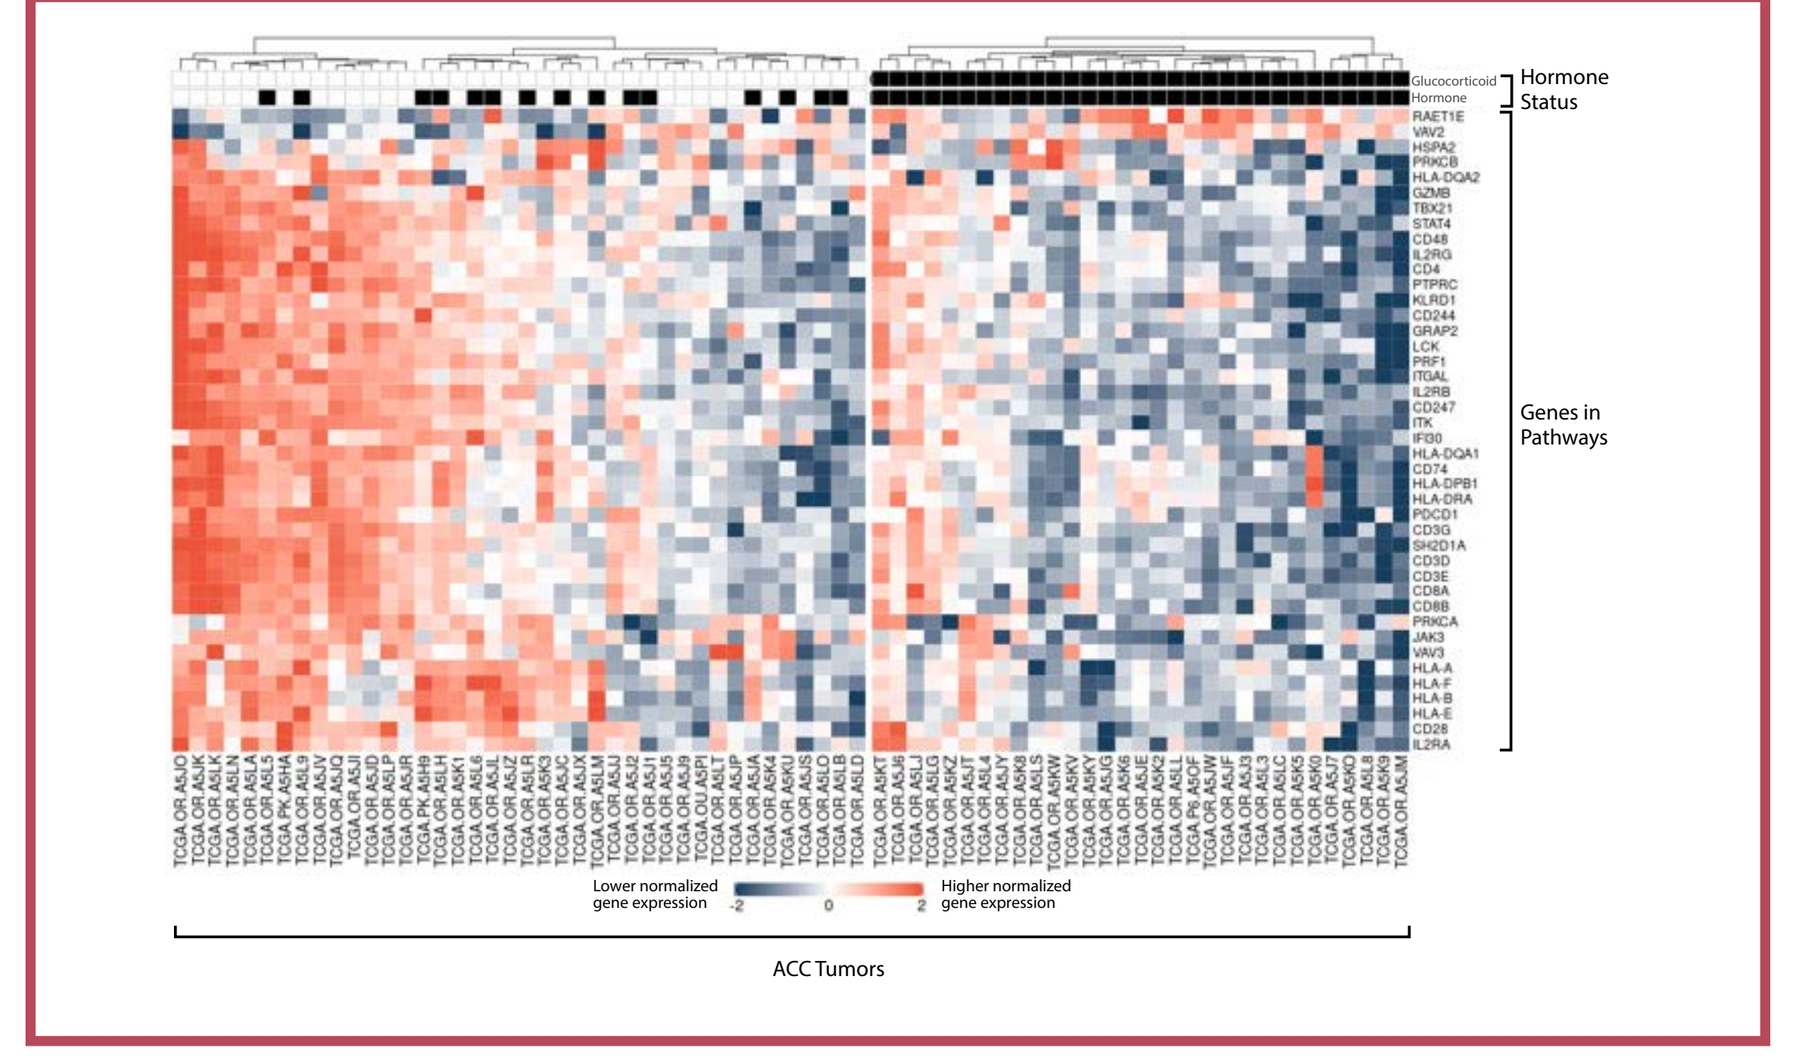
Hormone Status
Genes in Pathways
Lower normalized gene expression
Higher normalized gene expression
ACC Tumors

GC EXCESS IS ASSOCIATED WITH HYPOMETHYLATION OF STEROID SYNTHESIS GENES

- In GC+ ACC cases, a large number of genes were significantly hypomethylated (**Figure 6**, light blue), while fewer genes were hypermethylated (**Figure 6**, brown).
- Differences in methylation may explain the upregulation of steroidogenesis pathways but not the downregulation of immune pathways.
 - The hypomethylated genes were primarily associated with aldosterone, GC, and bile synthesis/secretion—pathways that are upregulated in GC+ ACC (**Figure 3**).
 - In contrast, the immune pathways with downregulated gene expression identified by mRNA analysis were not enriched in either the hypo- or hypermethylated sets.

Figure 6. Effects of GC on Promoter Methylation. More genes were significantly hypomethylated (*P*≤.05, Δ beta<-0.2) than hypermethylated (*P*≤.05, Δ beta>0.2) in GC+ tumors. Beta values represent the percentage of methylation in a gene.

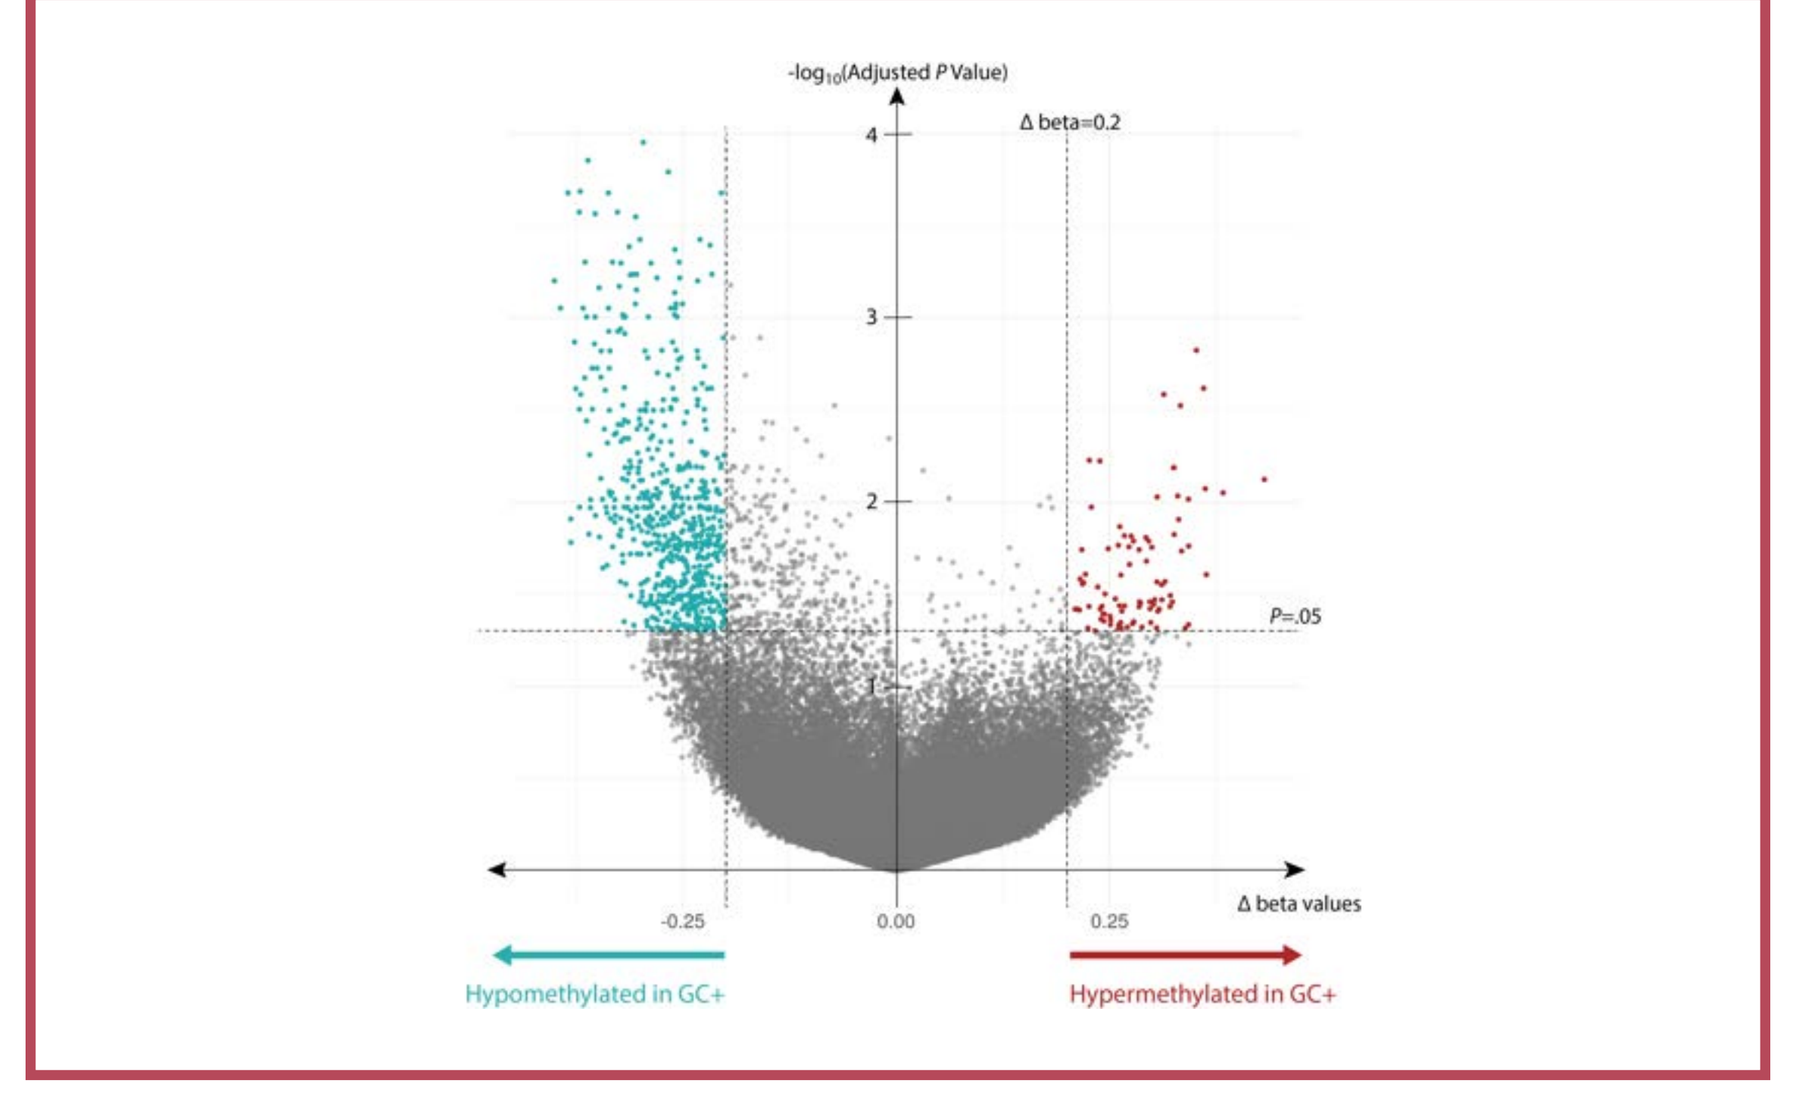
-log10(Adjusted P Value)
Δ beta=0.2
P=.05
Δ beta values
Hypomethylated in GC+
Hypermethylated in GC+

GC+ ACC TUMORS SHOW LOWER LYMPHOCYTE ABUNDANCE, HIGHER MYELOID AND MESENCHYMAL STEM CELL ABUNDANCE, AND HIGHER TUMOR MUTATION BURDEN

- Higher tumor mutation burden was also observed in the GC+ cases (*P*=.029, **Figure 7**).
- xCell analysis showed that T cells (*P*<.005) and natural killer T cells (NKT cells, *P*=.014) were less abundant in GC+ cases compared to GC- (**Figure 8**, top).
- In contrast, mesenchymal stem cells and neutrophils were more abundant in GC+ cases (*P*<.001, **Figure 8**, bottom).

Figure 7. Elevated Tumor Mutation Burden in GC+ ACC. In GC+ cases, more missense and nonsense mutations were observed compared to GC-.

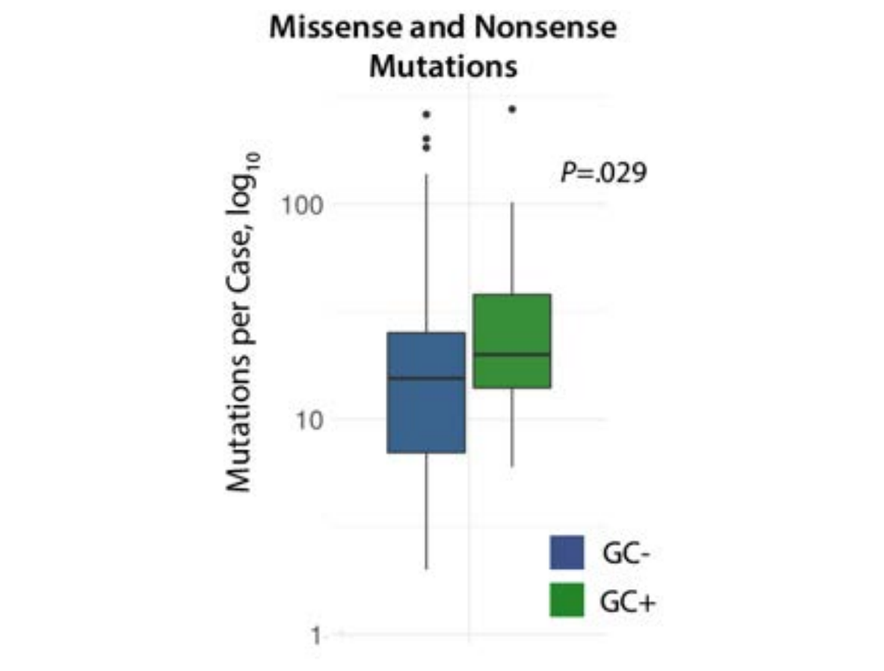
Missense and Nonsense Mutations
Mutations per Case, log10
P=.029
GC-
GC+

Figure 8. xCell Analysis of Cell Abundance in ACC Tumors. Lymphocyte abundance was lower (top), while mesenchymal stem cell and neutrophil abundances were higher (bottom) in GC+ cases.

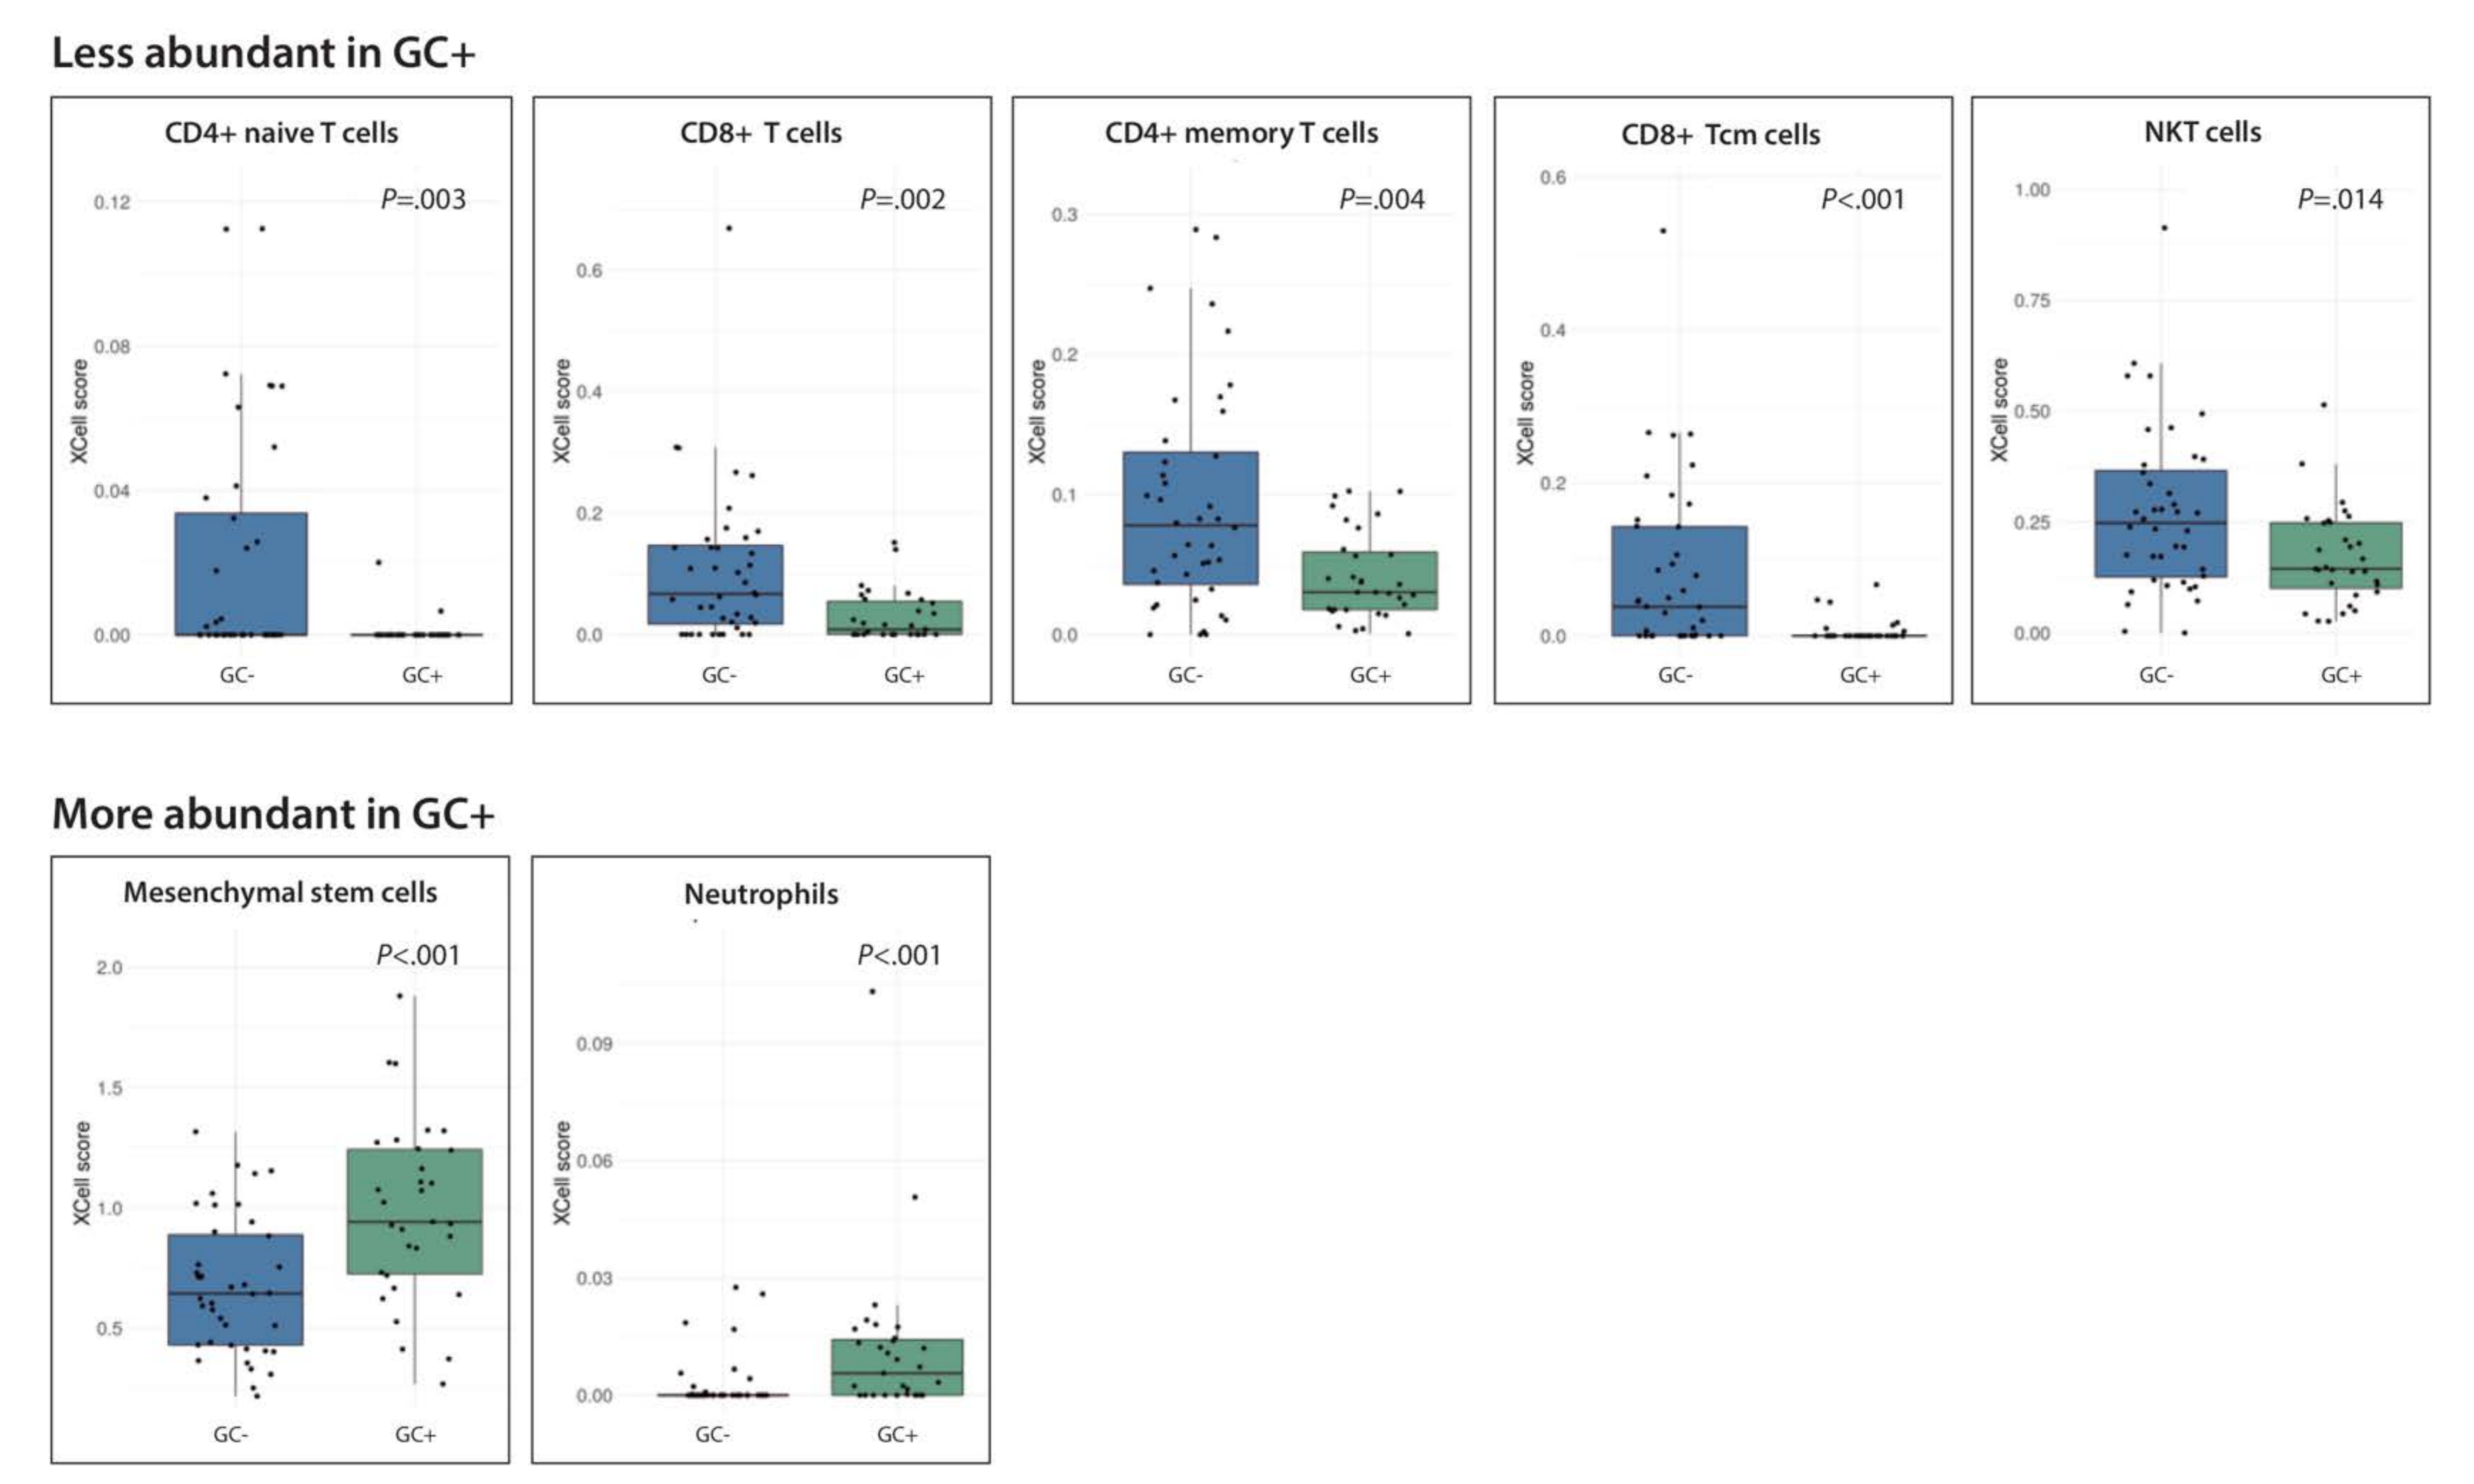
Less abundant in GC+
CD4+ naive T cells
P=.003
CD8+ T cells
P=.002
CD4+ memory T cells
P=.004
CD8+ Tcm cells
P<.001
NKT cells
P=.014
More abundant in GC+
Mesenchymal stem cells
P<.001
Neutrophils
P<.001
XCell score
GC-
GC+

GR ACTIVITY IS HIGH IN ACC, INDEPENDENT OF HORMONE STATUS

- Tumor scoring using a published GR-driven gene signature [14] confirmed that GR activity is high in ACC compared to other tumor types in TCGA (**Figure 9**).
- There was no difference between ACC cases with different hormone and GC status (insert in **Figure 9**).

Figure 9. GR Activity Score for Different Tumor Types and ACC Subsets. ACC exhibited high GR-driven gene activity relative to other tumors and independent of hormone status (see insert).

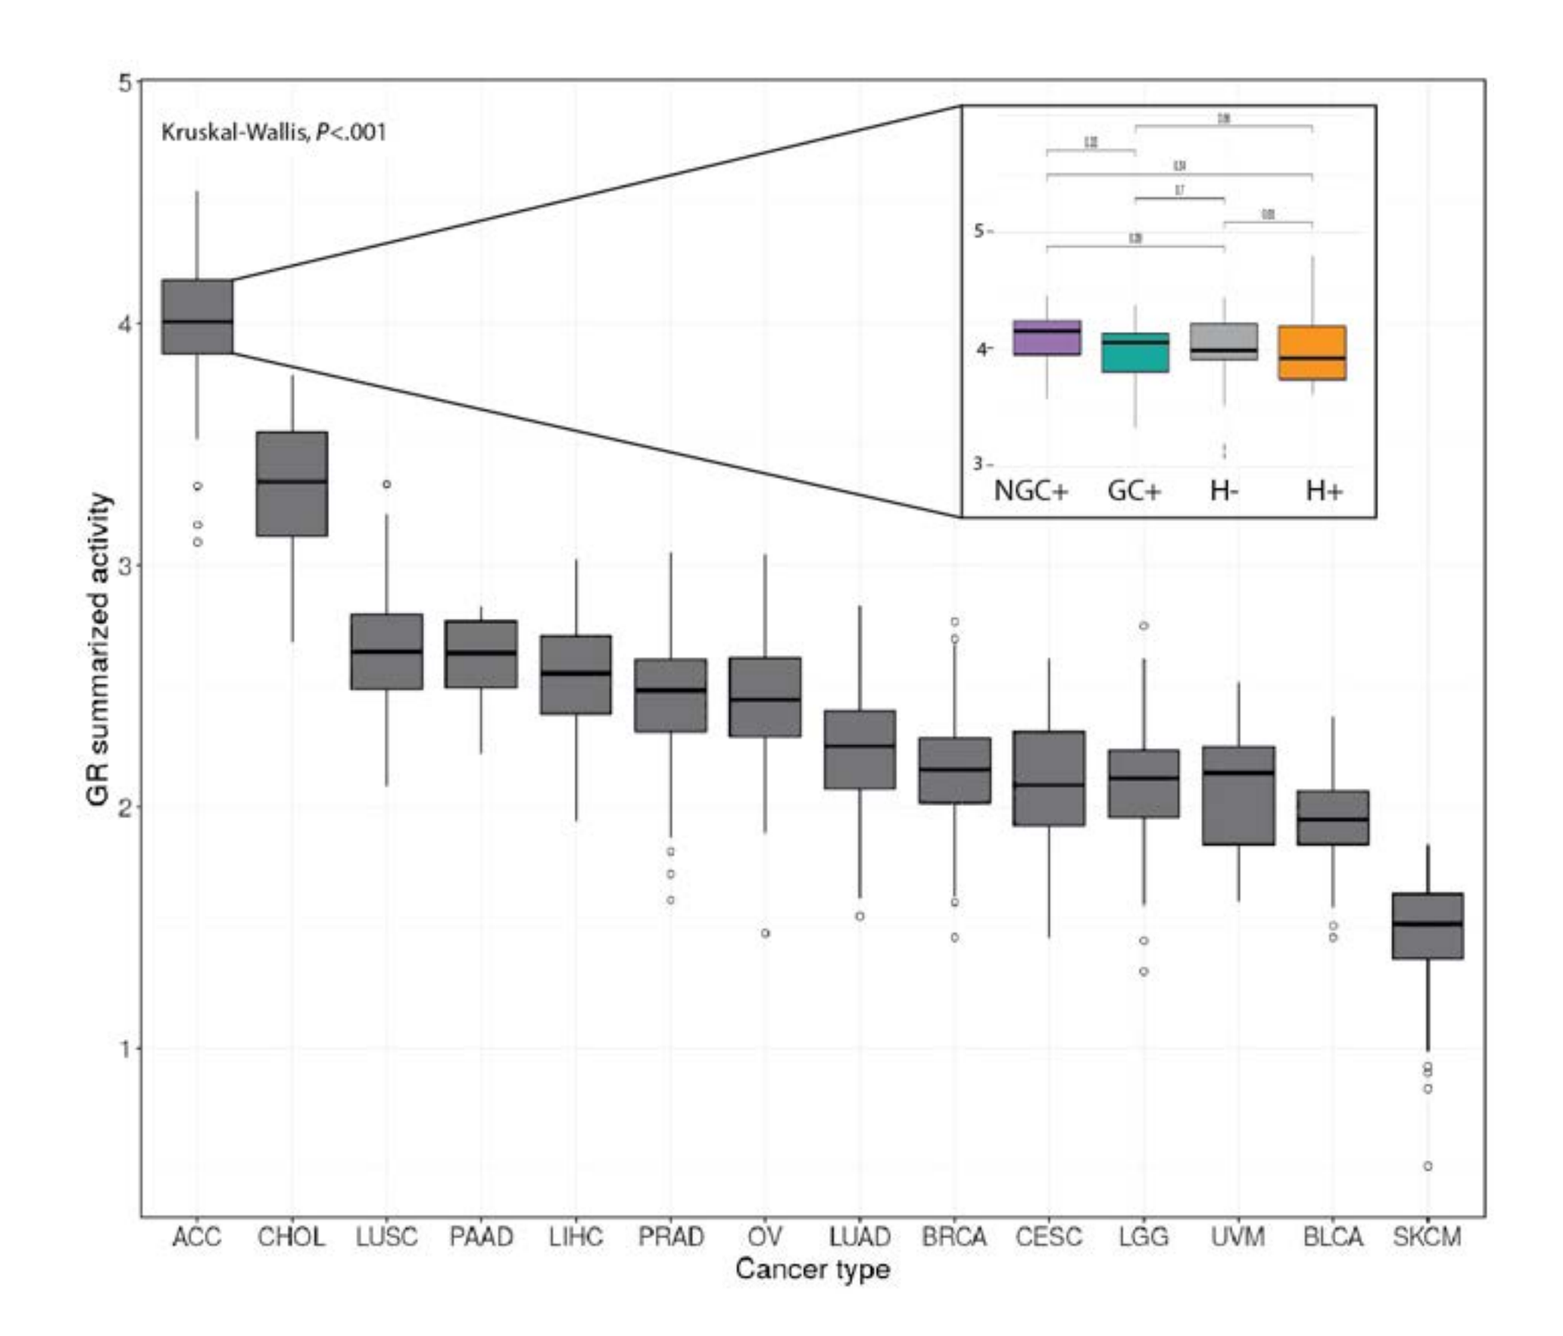
Kruskal-Wallis, P<.001
GR summarized activity
NGC+
GC+
H-
H+
ACC
CHOL
LUSC
PAAD
LIHC
PRAD
OV
LUAD
BRCA
CESC
LGG
UVM
BLCA
SKCM
Cancer type

GENE SIGNATURE CAN PREDICT GC+ LIKE TUMOR CASES

- Random forest methods were used to derive a model that distinguishes GC+/- ACC cases with ROC AUC = 0.87 ± 0.09 (**Figure 10**).
 - The sensor component of the inflammasome (NLRP1) and a mediator of NK activation by IL-15 (ZNF683) were identified as important parts of this signature (insert in **Figure 10**).

Figure 10. Derivation of a Gene Signature that Distinguishes GC+/- ACC Cases Using Random Forest. NLRP1 and ZNF683 (highlighted) were identified as important components of the signature. Only signature genes above the threshold of 0.0028 are shown.

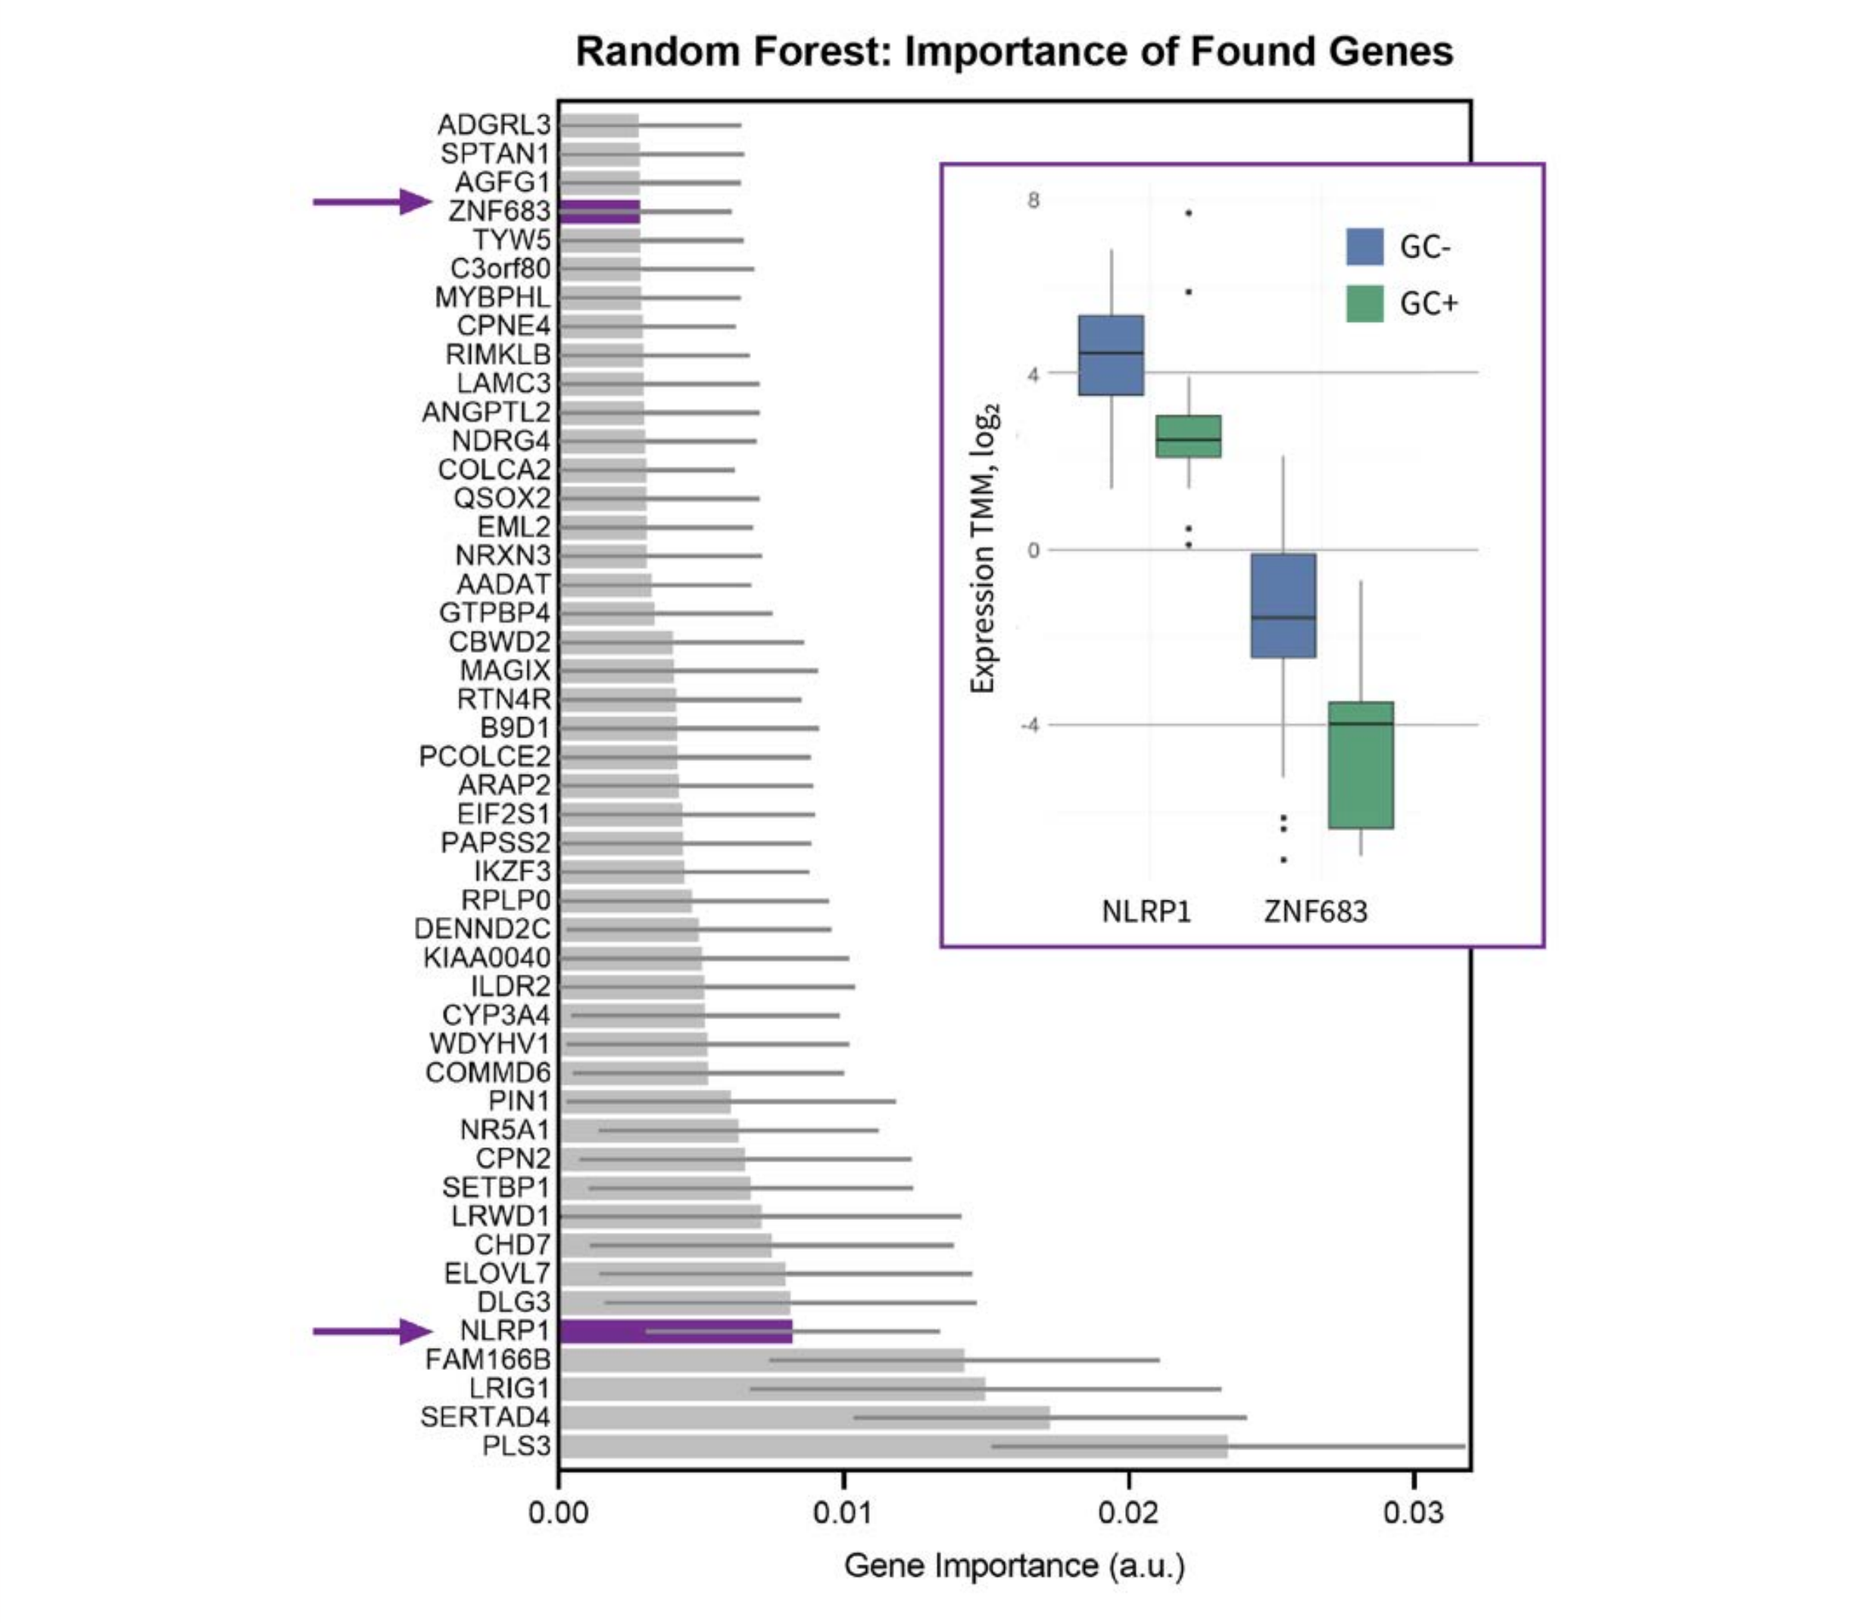
Random Forest: Importance of Found Genes
ADGRL3
SPTAN1
AGFG1
ZNF683
TYW5
C3orf80
MYBPHL
CPNE4
RIMKLB
LAMC3
ANGPTL2
NDRG4
COLCA2
QSOX2
EML2
NRXN3
AADAT
GTPBP4
CBWD2
MAGIX
RTN4R
B9D1
PCOLCE2
ARAP2
EIF2S1
PAPSS2
IKZF3
RPLP0
DENND2C
KIAA0040
ILDR2
CYP3A4
WDYHV1
COMMD6
PIN1
NR5A1
CPN2
SETBP1
LRWD1
CHD7
ELOVL7
DLG3
NLRP1
FAM166B
LRIG1
SERTAD4
PLS3
Gene Importance (a.u.)
Expression TMM, log2
GC-
GC+
NLRP1
ZNF683

- The gene signature was then applied to other tumors types in TCGA to identify those with GC+ like transcriptional profiles (**Figure 11**).
 - Based on the known distribution of GC+/- cases in ACC, a cutoff score of 0.75 was derived to distinguish GC+/- tumors (horizontal line in **Figure 11A**).
 - According to this score, uveal (UVM) and skin cutaneous melanomas (SKCM) may have the highest frequency of cases similar to GC+ ACC (**Figure 11B**).

Figure 11. Application of the ACC Gene Signature to TCGA Tumors (A) and Predicted Frequency of Tumor Cases Resembling GC+ ACC (B). Uveal (UVM) and skin cutaneous melanomas (SKCM) are predicted to have the highest frequency of tumors that resemble GC+ ACC.

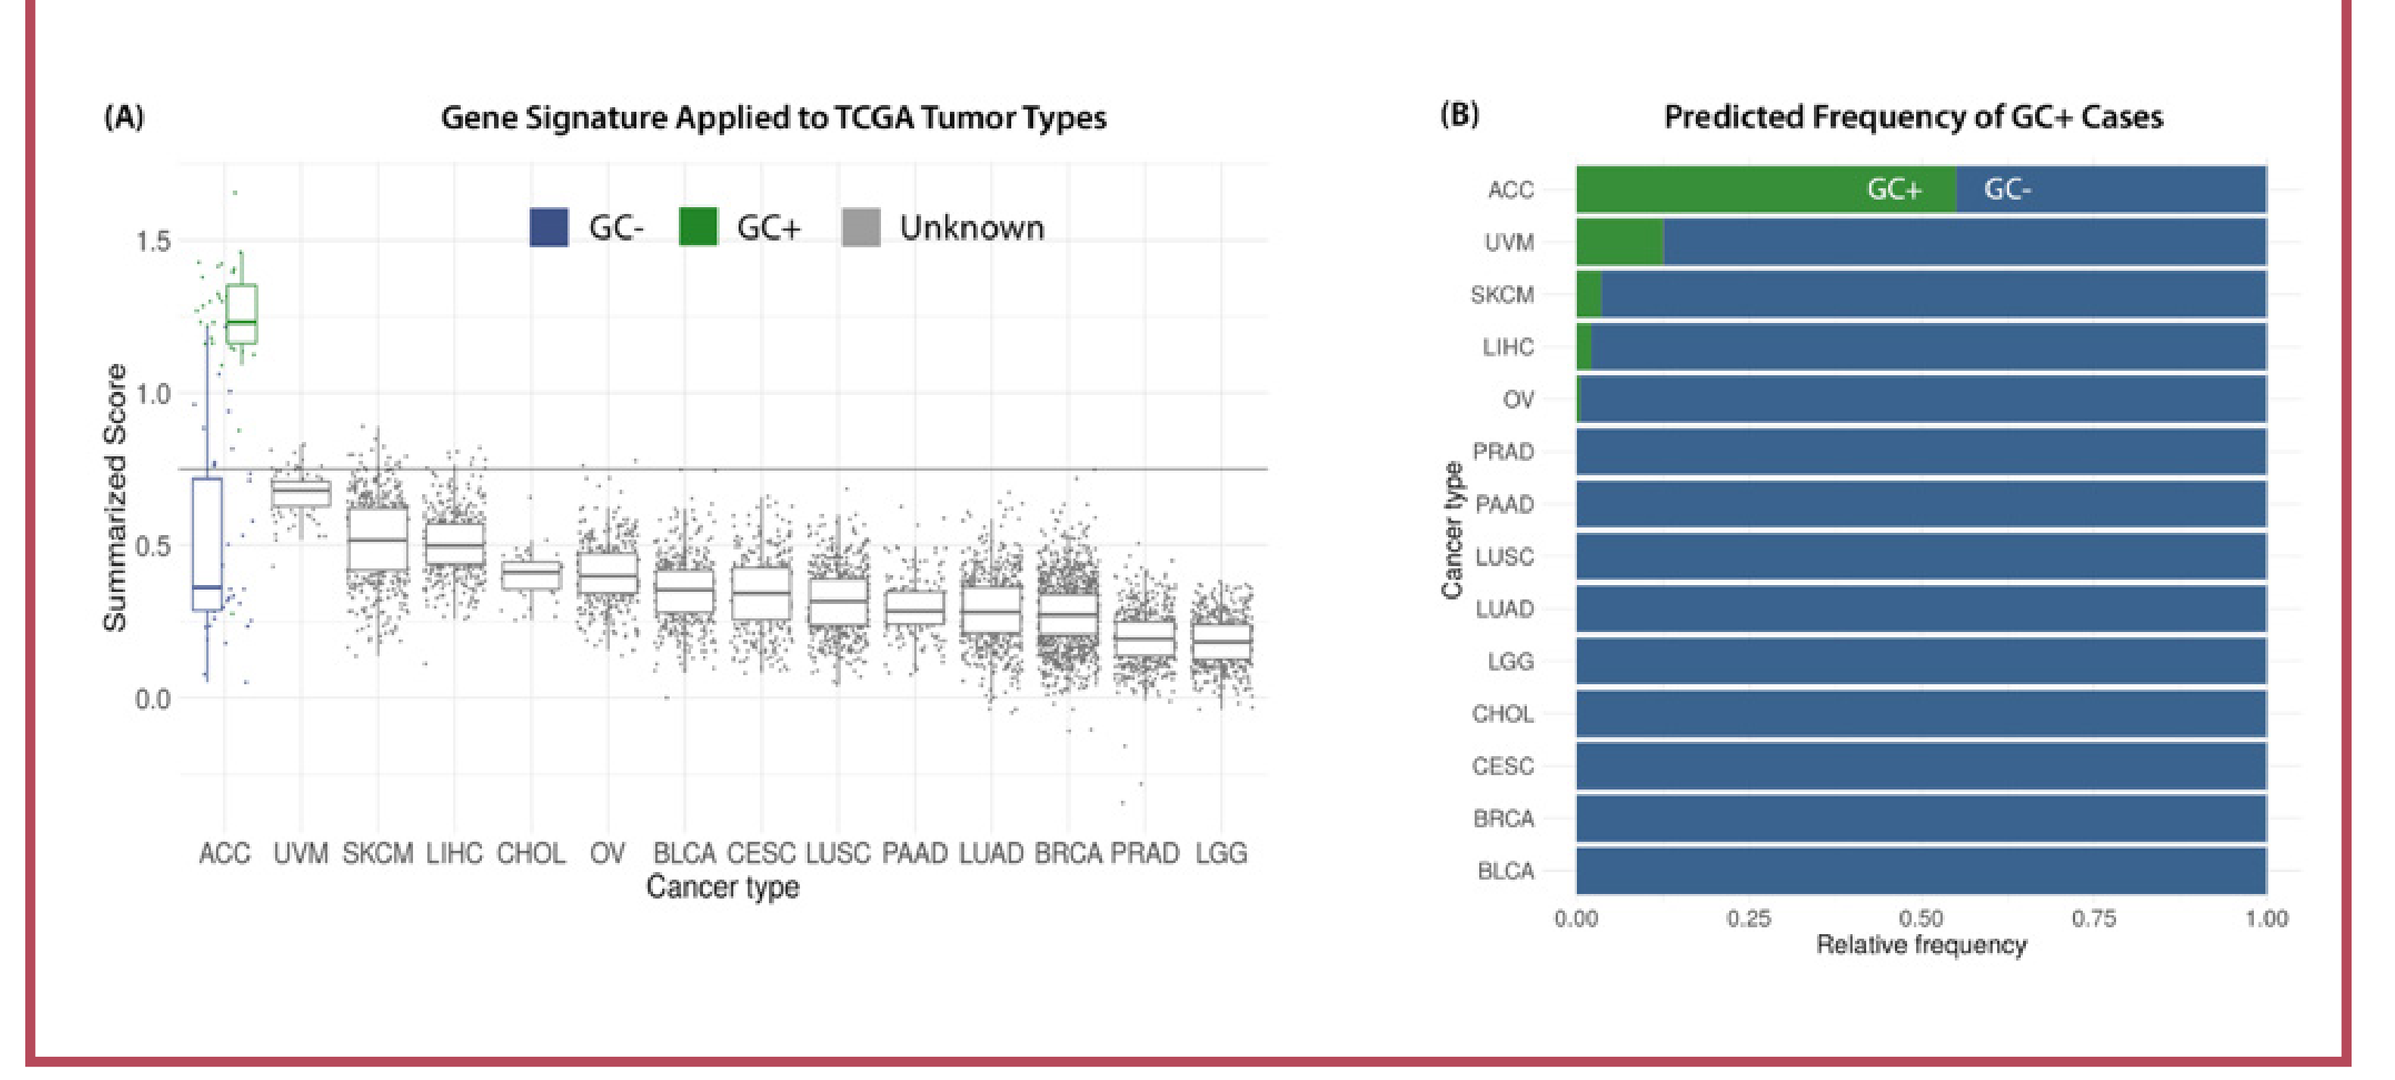
(A)
Gene Signature Applied to TCGA Tumor Types
GC-
GC+
Unknown
Summarized Score
ACC UVM SKCM LIHC CHOL OV BLCA CESC LUSC PAAD LUAD BRCA PRAD LGG
Cancer type
(B)
Predicted Frequency of GC+ Cases
Cancer type
Relative frequency

CONCLUSIONS

- Glucocorticoid excess affects significantly more genes in ACC than other hormones.
- In GC+ ACC, expression of steroid synthesis genes was elevated while immune-related genes were suppressed.
 - Normal adrenal cells express steroid synthesis genes but not immune-related genes.
 - Steroid synthesis genes were hypomethylated in GC+ cases, while no difference in methylation between GC+ and GC- cases was found for immune genes.
 - Furthermore, fewer infiltrating immune cells (T cells and NKT cells) were found in GC+ cases compared to GC-, suggesting that immune effects are due to changes in immune cell infiltrate rather than transcriptional changes.
- Tumor mutation burden was higher in GC+ ACC cases, which may be caused by the observed immune suppression or immune cell exclusion that may, in turn, be related to higher tolerance of non-self-antigens in GC+ cases.
- A published GR activity score showed no difference between GC+ and GC- cases, which may be due to locally high concentrations of GC in the adrenal gland regardless of systemic GC levels.
 - In contrast, immune infiltration into ACC tumors may be negatively affected by the exposure of lymph nodes to elevated GC activity.
- A newly derived gene signature predicts the highest frequency of GC+-like tumors in uveal and skin cutaneous melanomas.
- The observed reduced abundance of immune cells and immune-related transcripts in GC+ ACC provides insight into the mechanisms by which GC may limit response to ICI therapy.
 - GR antagonism may increase immune related transcripts or immune cell infiltration, thus promoting tumor immune response in GC+ ACC and other malignancies with elevated GC activity.
 - This hypothesis will be tested in a phase 1 oncology trial of relacorilant combined with ICI (CORT125134-551, NCT04373265).

REFERENCES

1. Abercrombie, H.C., et al. *Psychoneuroendocrinology*, 2004. 29(8): p. 1082-92.
2. Mormont, M.C. and F. Levi. *Int J Cancer*, 1997. 70(2): p. 241-7.
3. Sharma, P., et al. *J Oral Maxillofac Pathol*, 2018. 22(1): p. 27-34.
4. Palesh, O., et al. *J Clin Sleep Med*, 2008. 4(5): p. 441-9.
5. Jehn, C.F., et al. *Integr Cancer Ther*, 2010. 9(3): p. 270-5.
6. Arbour, K.C., et al. *J Clin Oncol*, 2018. 36(28): p. 2872-2878.
7. Scott, S.C. and N.A. Pennell. *J Thorac Oncol*, 2018. 13(11): p. 1771-1775.
8. Fuca, G., et al. *ESMO Open*, 2019. 4(1): p. e000457.
9. Else, T., et al. *J Clin Endocrinol Metab*, 2014. 99(2): p. 455-61.
10. Habra, M.A., et al. *J Immunother Cancer*, 2019. 7(1): p. 253.
11. Raj, N., et al. *J Clin Oncol*, 2020. 38(1): p. 71-80.
12. Zheng, S., et al. *Cancer Cell*, 2016. 30(2): p. 363.
13. Aran, D., Z. Hu, and A.J. Butte. *Genome Biol*, 2017. 18(1): p. 220.
14. West, D.C., et al. *Clin Cancer Res*, 2018. 24(14): p. 3433-3446.

ACKNOWLEDGEMENTS

The results shown here are based upon data generated by the TCGA Research Network: https://www.cancer.gov/tcga.
Editorial support was provided by Tina K. Schlafly of Corcept Therapeutics. Funding for design and production support for this poster was provided by Corcept to MedVal Scientific Information Services, LLC (Princeton, NJ). The authors developed and revised the poster and provided approval of the final version.

DISCLOSURES

AEG and AG: Employee, Corcept; **SPS:** Former employee, Corcept; Stock ownership in Corcept, Abbot, and AbbVie; **MAH:** Consultant/Advisor, Corcept, HRA Pharma, and Calico; Investigator/Researcher for Exelixis, **MM, PB, MJP:** Employee, Ardigen

Supported by funding from Corcept Therapeutics

Presented at the American Association for Cancer Research (AACR) Virtual Annual Meeting II, June 22-24, 2020